Update on the INSPIRE DUCHENNE Phase 1/2 Study of SGT-003 Microdystrophin Gene Therapy

Eugenio Mercuri[1], Aravindhan Veerapandiyan[2], Kevin Flanigan[3], Craig McDonald[4], Crystal Proud[5], Perry Shieh[6], Hernan Gonorazky[7], Alicia Henriquez[8], Craig Zaidman[9], Nancy Kuntz[10], Renata Shih[11], Erika Finanger[12], Jamie Marshall[13], Nicolas Christoforou[13], Jessie Hanrahan[13], Patrick Gonzalez[13], Matthew Harmelink[13], Gabriel Brooks[13]

1. Fondazione Policlinico Gemelli, Rome, Italy; 2. Arkansas Children's Hospital, Little Rock, AR, USA; 3. Nationwide Children's Hospital, Columbus, OH, USA; 4. University of California, Davis, Sacramento, CA, USA; 5. Children's Hospital of the King's Daughters, Norfolk, VA, USA; 6. University of California, Los Angeles, Los Angeles, CA, USA; 7. The Hospital for Sick Children, Toronto, ON, Canada; 8. Seattle Children's Hospital, Seattle, WA, USA; 9. Washington University at St. Louis, St Louis, MO, USA; 10. Lurie Children's Hospital, Chicago, IL, USA; 11. Rare Disease Research, Atlanta, GA, USA; 12. Oregon Health and Science University, Portland, OR, USA; 13. Solid Biosciences Inc., Charlestown, MA, USA

SOLID BIOSCIENCES

INTRODUCTION

Background

- Duchenne muscular dystrophy (Duchenne) is a chronic, progressive, and ultimately fatal X-linked recessive neuromuscular disorder caused by the lack of functional dystrophin protein[1]
 - Dystrophin is required for maintaining muscle integrity and function[2-4]
 - Deterioration of muscle integrity leads to muscle fiber breakdown, resulting in progressive functional decline
- Shortened, functional microdystrophin transgenes can be packaged into adeno-associated viruses (AAVs) to replace dystrophin[5]
 - Microdystrophins vary based on their unique compositions[6]

SGT-003 Overview

- SGT-003 is Solid's next-generation AAV-microdystrophin gene therapy with key features:
 - Microdystrophin transgene: unique inclusion of the R16/R17 neuronal nitric oxide synthase (nNOS)-binding domain with the goal of preventing activity-induced ischemia and associated muscle injury[7]

SGT-003 MICRODYSTROPHIN TRANSGENE

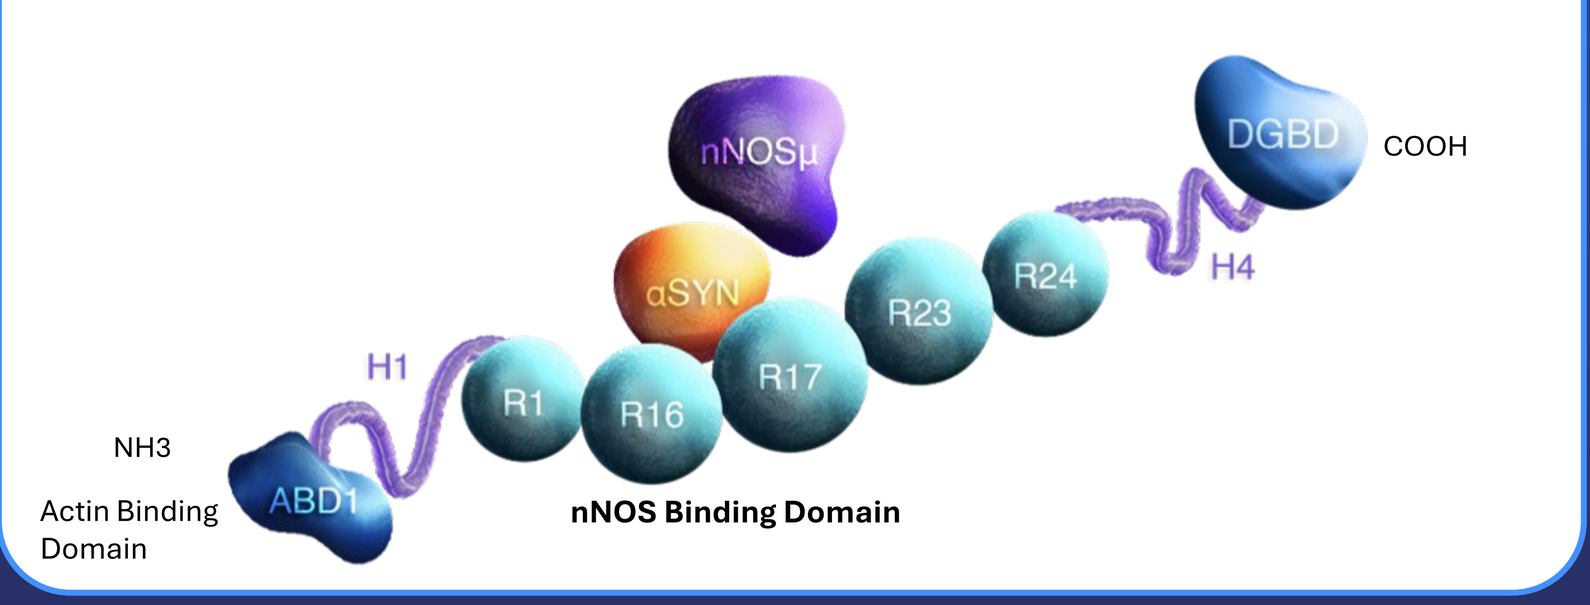
nNOSμ
DGBD
COOH
αSYN
R24
R23
H4
H1
R1
R16
R17
NH3
Actin Binding Domain
ABD1
nNOS Binding Domain

 - POLARIS-101™ capsid (formerly known as AAV-SLB101): rationally designed muscle-tropic capsid that leads to higher levels in muscle and reduced levels in the liver[8]
 - Full/empty capsid ratio: SGT-003 manufacturing uses a transient transfection process, focusing on maintaining high % full capsid ratios to improve purity

INSPIRE DUCHENNE (NCT06138639) Study Design

- Phase 1/2, open-label study evaluating the safety, tolerability, and efficacy of a single intravenous (IV) infusion of SGT-003 at 1E14 vg/kg
- Actively enrolling participants 0 to < 12 years of age with a confirmed diagnosis of Duchenne
- Uses a steroid-only regimen for prophylactic immunomodulation
- Primary safety endpoint: incidence of treatment-emergent adverse events (TEAEs) through Day 360
- Primary efficacy endpoint: change from baseline of microdystrophin protein levels at Day 90
- Secondary endpoints include:
 - Microdystrophin expression: Protein levels at Day 360 and distribution at Day 90 and Day 360
 - Motor function: Time-to-Rise, 10-Meter Walk/Run, North Star Ambulatory Assessment, 4-Stair Climb, 6-Minute Walk Test, Stride Velocity 95th Centile

RESULTS

SGT-003 HAS BEEN GENERALLY WELL TOLERATED

- No drug-induced liver injury, thrombotic microangiopathy, atypical hemolytic uremic syndrome, or myocarditis have been observed to date

Table 1. INSPIRE DUCHENNE Enrollment[9]

Cohorts	Eligible age range (years)	Ages at enrollment (years)	Weights for dosing (kg)	Participants enrolled (n)
1-3	0 to <12	6 mo. to 10 years	6.5 to 39.7	50/53*

*Participants 51-53 were dosed after the safety data cutoff of May 29, 2026. Solid Biosciences. 2026.

Table 2. INSPIRE DUCHENNE Safety Data[10]

SGT-003 Participants With Treatment-Related SAEs	n (%)
Serious Adverse Events (SAEs)	2 (4.0)*

*One (n=1) participant with a possibly related transient adverse event of inflammation treated with two doses of antibiotics. One (n=1) previously reported immune mediated myositis. Both events have resolved.

SGT-003 Participants With Treatment-Related AEs		n (%)
Most Common Adverse Events (AEs)	Vomiting	32 (64.0)
	Nausea	27 (54.0)
	Decreased Appetite	15 (30.0)
	Thrombocytopenia	13 (26.0)
	Abdominal Pain	9 (18.0)

Data cutoff of May 29, 2026. Solid Biosciences.

REFERENCES

1. Duan D, et al. *Nat Rev Dis Primers*. 2021;7(1):13. **2.** Sheybani A, et al. *Pediatr Res*. 2022;92(6):1613-20. **3.** Voleti S, et al. *Pediatr Cardiol*. 2020;41(6):1173-9. **4.** Wagner KR, et al. *Biomark Med*. 2021;15(15):1389-96. **5.** Crudele JM, Chamberlain JS. *Hum Mol Genet*. 2019;28(R1):R102-R7. **6.** Ramos JN, et al. *Mol Ther*. 2019;27(3):623-35. **7.** Lai Y, et al. *J Clin Invest*. 2009;119(3):624-35. **8.** Vu Hong A, et al. *Nat Commun*. 2024;15(1):7965. **9.** Data on File. Data cutoff August 4, 2026. Solid Biosciences. **10.** Shieh P, et al. Presented at the International Congress on Neuromuscular Diseases, July 7-11, 2026, Florence, Italy. Data cutoff of May 29, 2026. **11.** Flanigan K, et al. Presented at ASGCT, May 5-11, 2026, Boston, MA. Data cutoff of February 23, 2026. **12.** "Baseline at or below normal" was defined as LVEF ≤60 at baseline: Romanowicz J, et al. J Am Soc Echocardiogr. 2023;36(3):310-323. **13.** For participants with low-normal baseline LVEF, increases in LVEF are beneficial and decreases unfavorable.

ACKNOWLEDGMENTS

The authors gratefully acknowledge the participants and their families for their participation and trust, as well as the dedication of the study site teams. This study is sponsored by Solid Biosciences Inc. (Charlestown, MA, USA).

RESULTS Cont'd

SGT-003 DEMONSTRATED ROBUST TRANSDUCTION & MICRODYSTROPHIN EXPRESSION

Table 3. Vector Genome Copies / Nucleus (Mean)[10]	**Day 90** (N=20)	**Day 360** (N=3)
	11	12

Figure 1. SGT-003 Microdystrophin Expression in INSPIRE DUCHENNE Muscle Biopsy Samples[11]

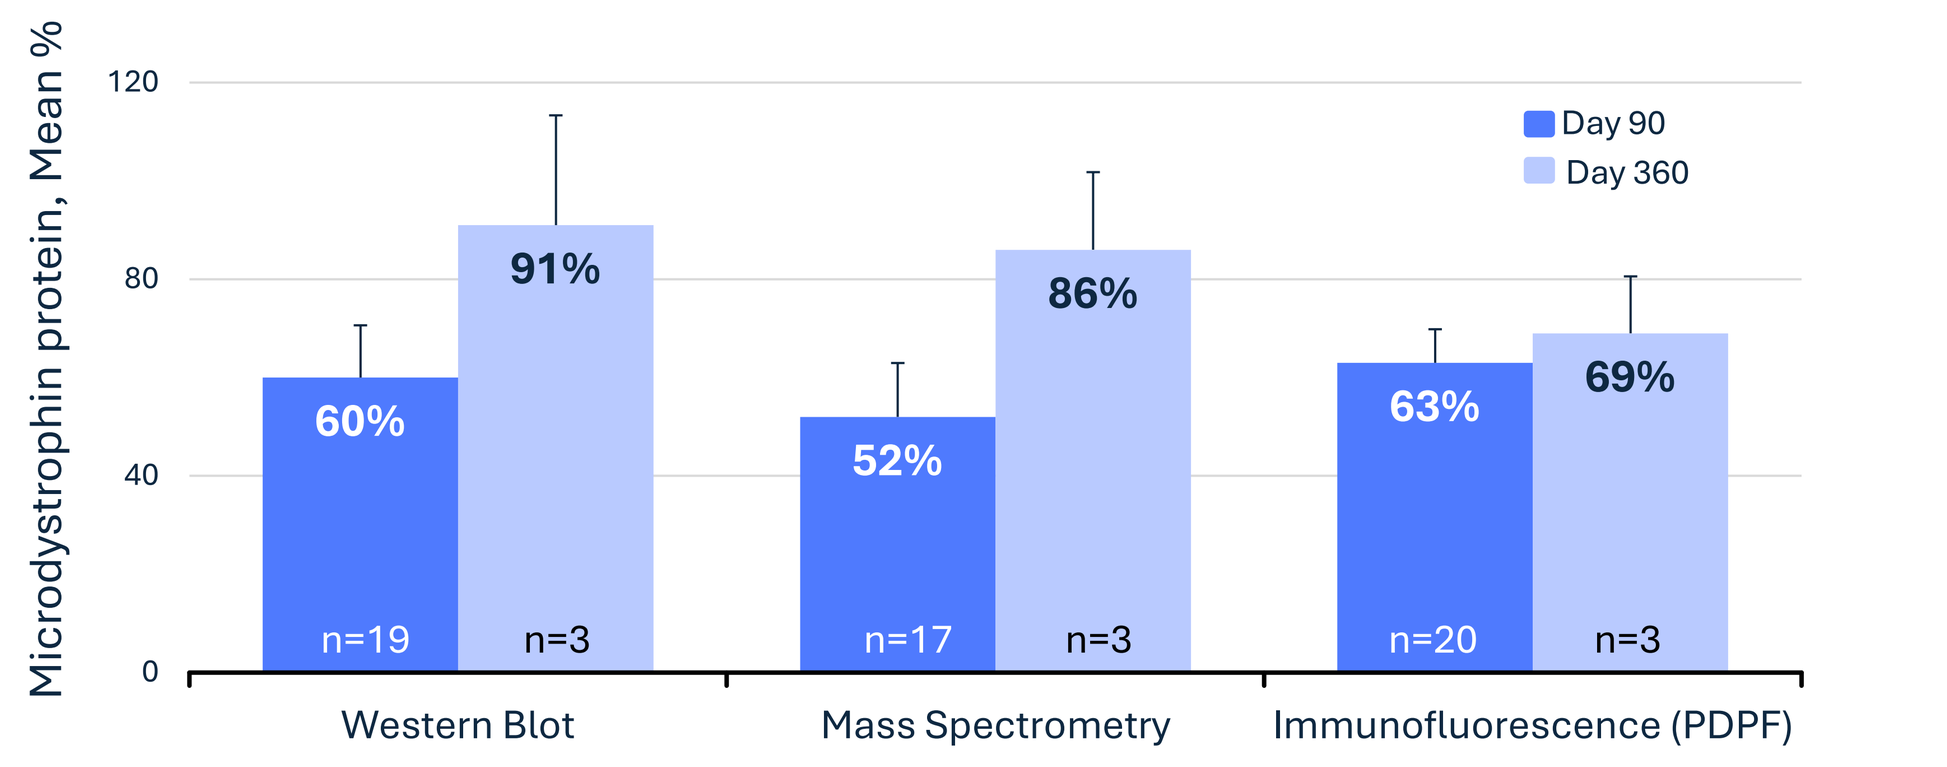
Microdystrophin protein, Mean %
120
80
40
0
Day 90
Day 360
60%
91%
52%
86%
63%
69%
n=19
n=3
n=17
n=3
n=20
n=3
Western Blot
Mass Spectrometry
Immunofluorescence (PDPF)

PDPF: percent dystrophin positive fibers

Figure 1. Values are means ± standard error of the mean. Western blot and mass spectrometry baselines were 0% mean normal dystrophin, immunofluorescence is based on a manual count. Western blot, mass spectrometry and immunofluorescence assays are conducted by multiple external vendors; at the time of this analysis, one western blot sample (from participant 20) and three mass spectrometry samples (from participants 15,16 and 20) had not been received

SGT-003 RESTORES KEY ELEMENTS OF THE DAPC

Figure 2. Immunofluorescence Evaluation of Dystrophin Associated Protein Complex (DAPC) Restoration in INSPIRE DUCHENNE Muscle Biopsy Samples[11]

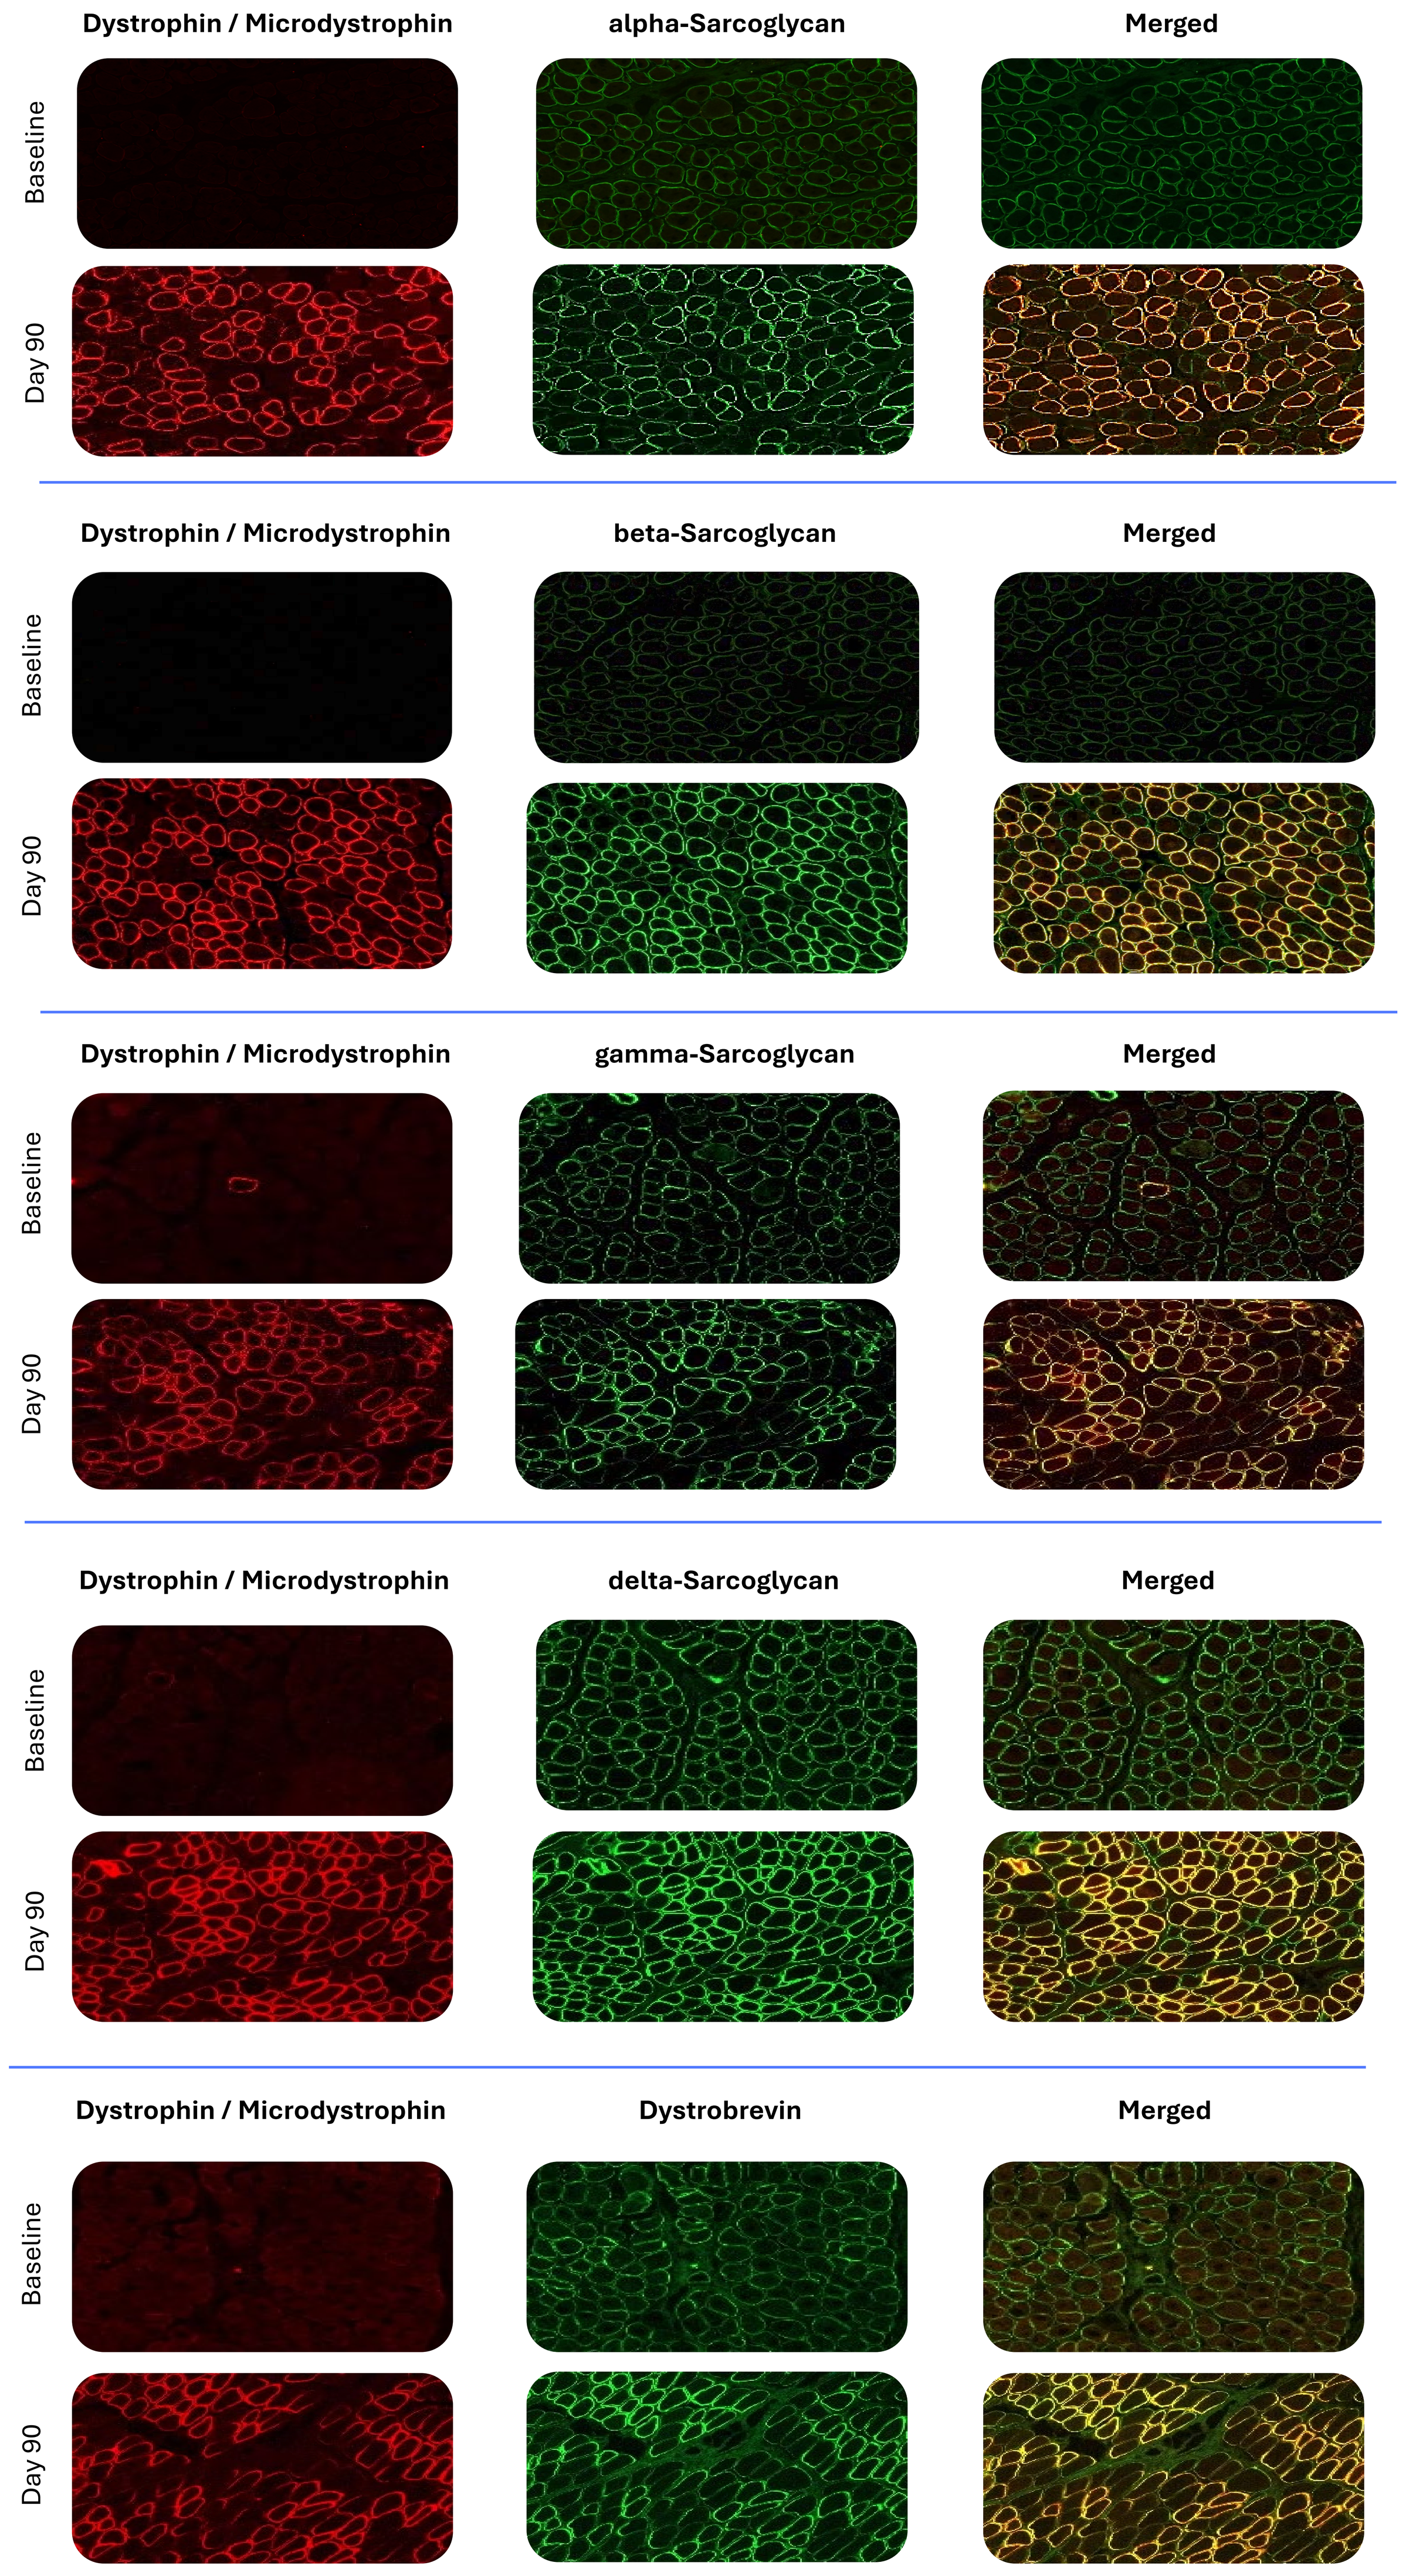
Dystrophin / Microdystrophin
alpha-Sarcoglycan
Merged
Baseline
Day 90
Dystrophin / Microdystrophin
beta-Sarcoglycan
Merged
Baseline
Day 90
Dystrophin / Microdystrophin
gamma-Sarcoglycan
Merged
Baseline
Day 90
Dystrophin / Microdystrophin
delta-Sarcoglycan
Merged
Baseline
Day 90
Dystrophin / Microdystrophin
Dystrobrevin
Merged
Baseline
Day 90

Figure 2. Muscle biopsies showed restored membrane localization of the DAPC, corresponding with microdystrophin expression. Representative images are shown from the same participant.

Figure 3. Quantification of nNOS & β-sarcoglycan Sarcolemmal Restoration in INSPIRE DUCHENNE Muscle Biopsy Samples[11]

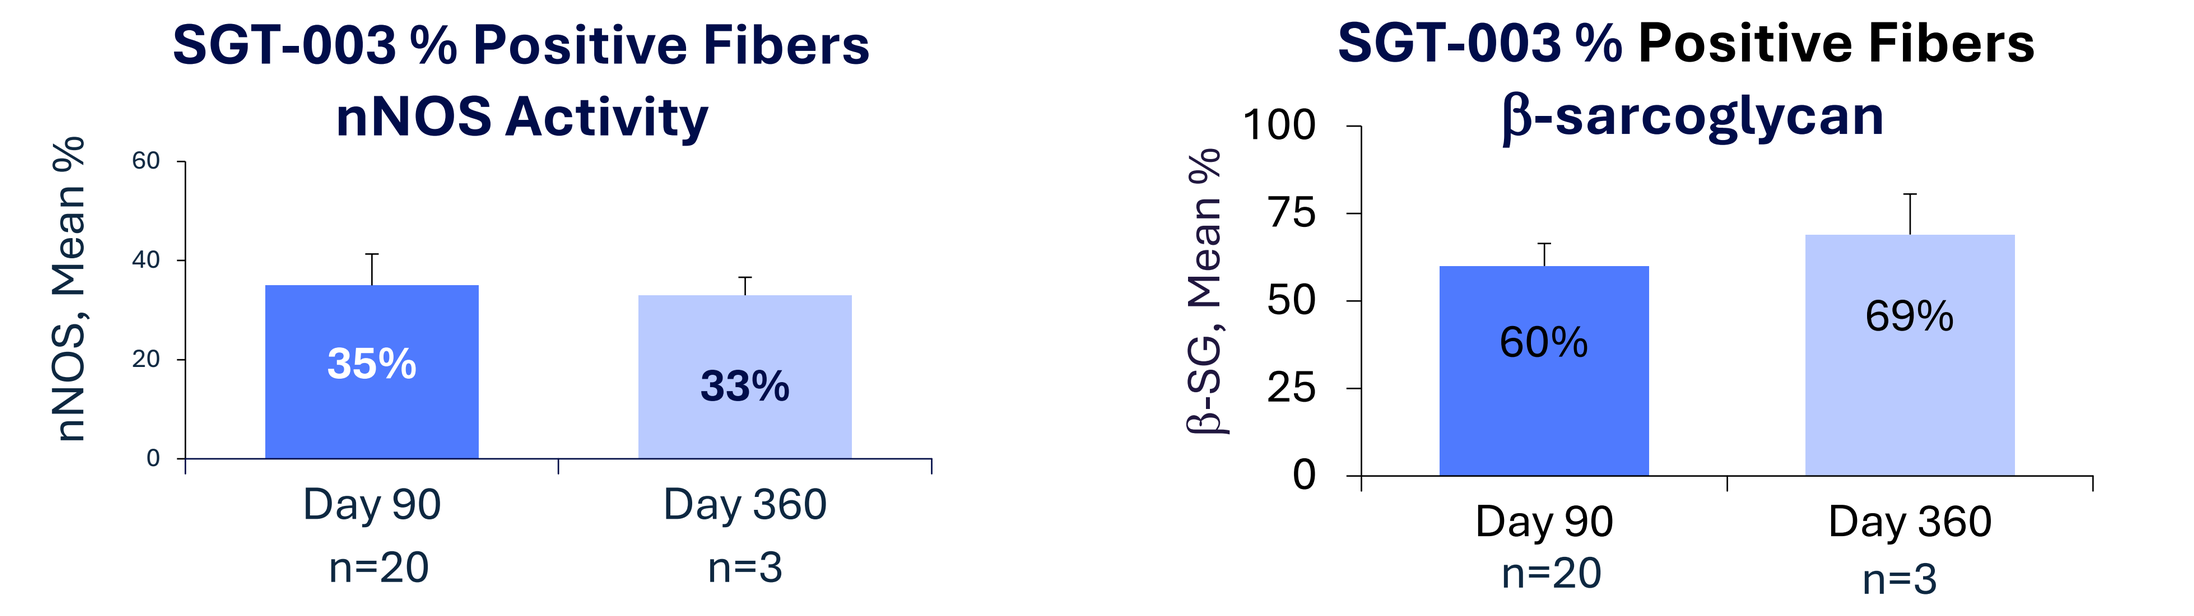
SGT-003 % Positive Fibers
nNOS Activity
nNOS, Mean %
60
40
20
0
35%
33%
Day 90
n=20
Day 360
n=3
SGT-003 % Positive Fibers
β-sarcoglycan
β-SG, Mean %
100
75
50
25
0
60%
69%
Day 90
n=20
Day 360
n=3

nNOS: neuronal nitric oxide synthase

Figure 3. Beta-sarcoglycan and nNOS values were based on a manual count.

SGT-003 IMPROVES MUSCLE HEALTH

Figure 4. Reduced Chronic Regenerative Demand Observed Post-SGT-003 Treatment[11]

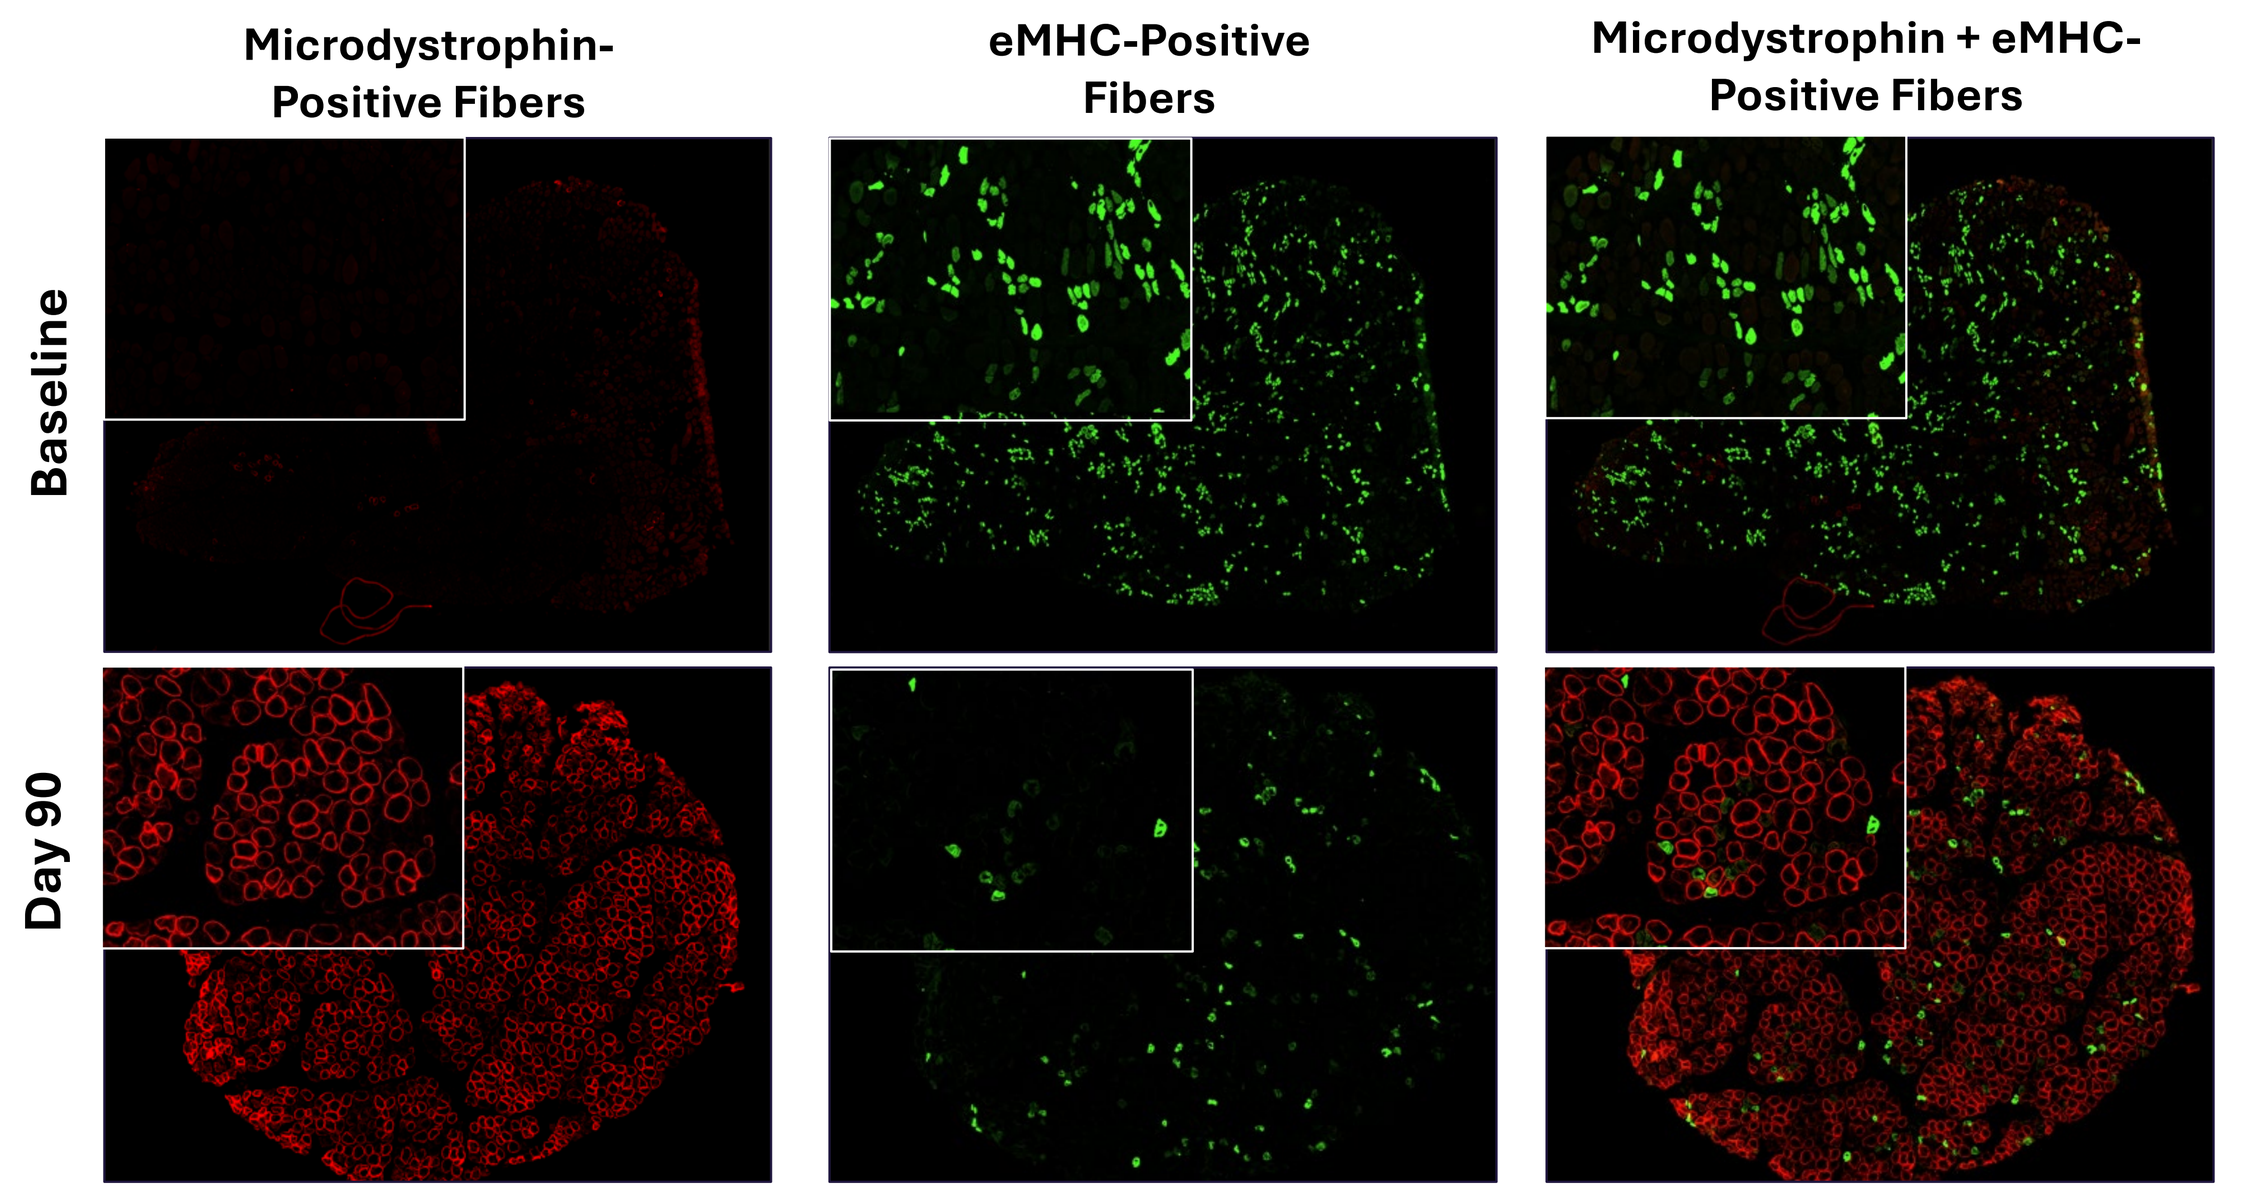
Microdystrophin-Positive Fibers
eMHC-Positive Fibers
Microdystrophin + eMHC-Positive Fibers
Baseline
Day 90

Figure 4. The reduction in eMHC suggests reduced need for muscle repair and preservation of muscle architecture.

SGT-003 SHOWS IMPROVED MUSCLE INTEGRITY

Figure 5. Reductions in Markers of Disrupted Muscle Integrity After SGT-003[11]

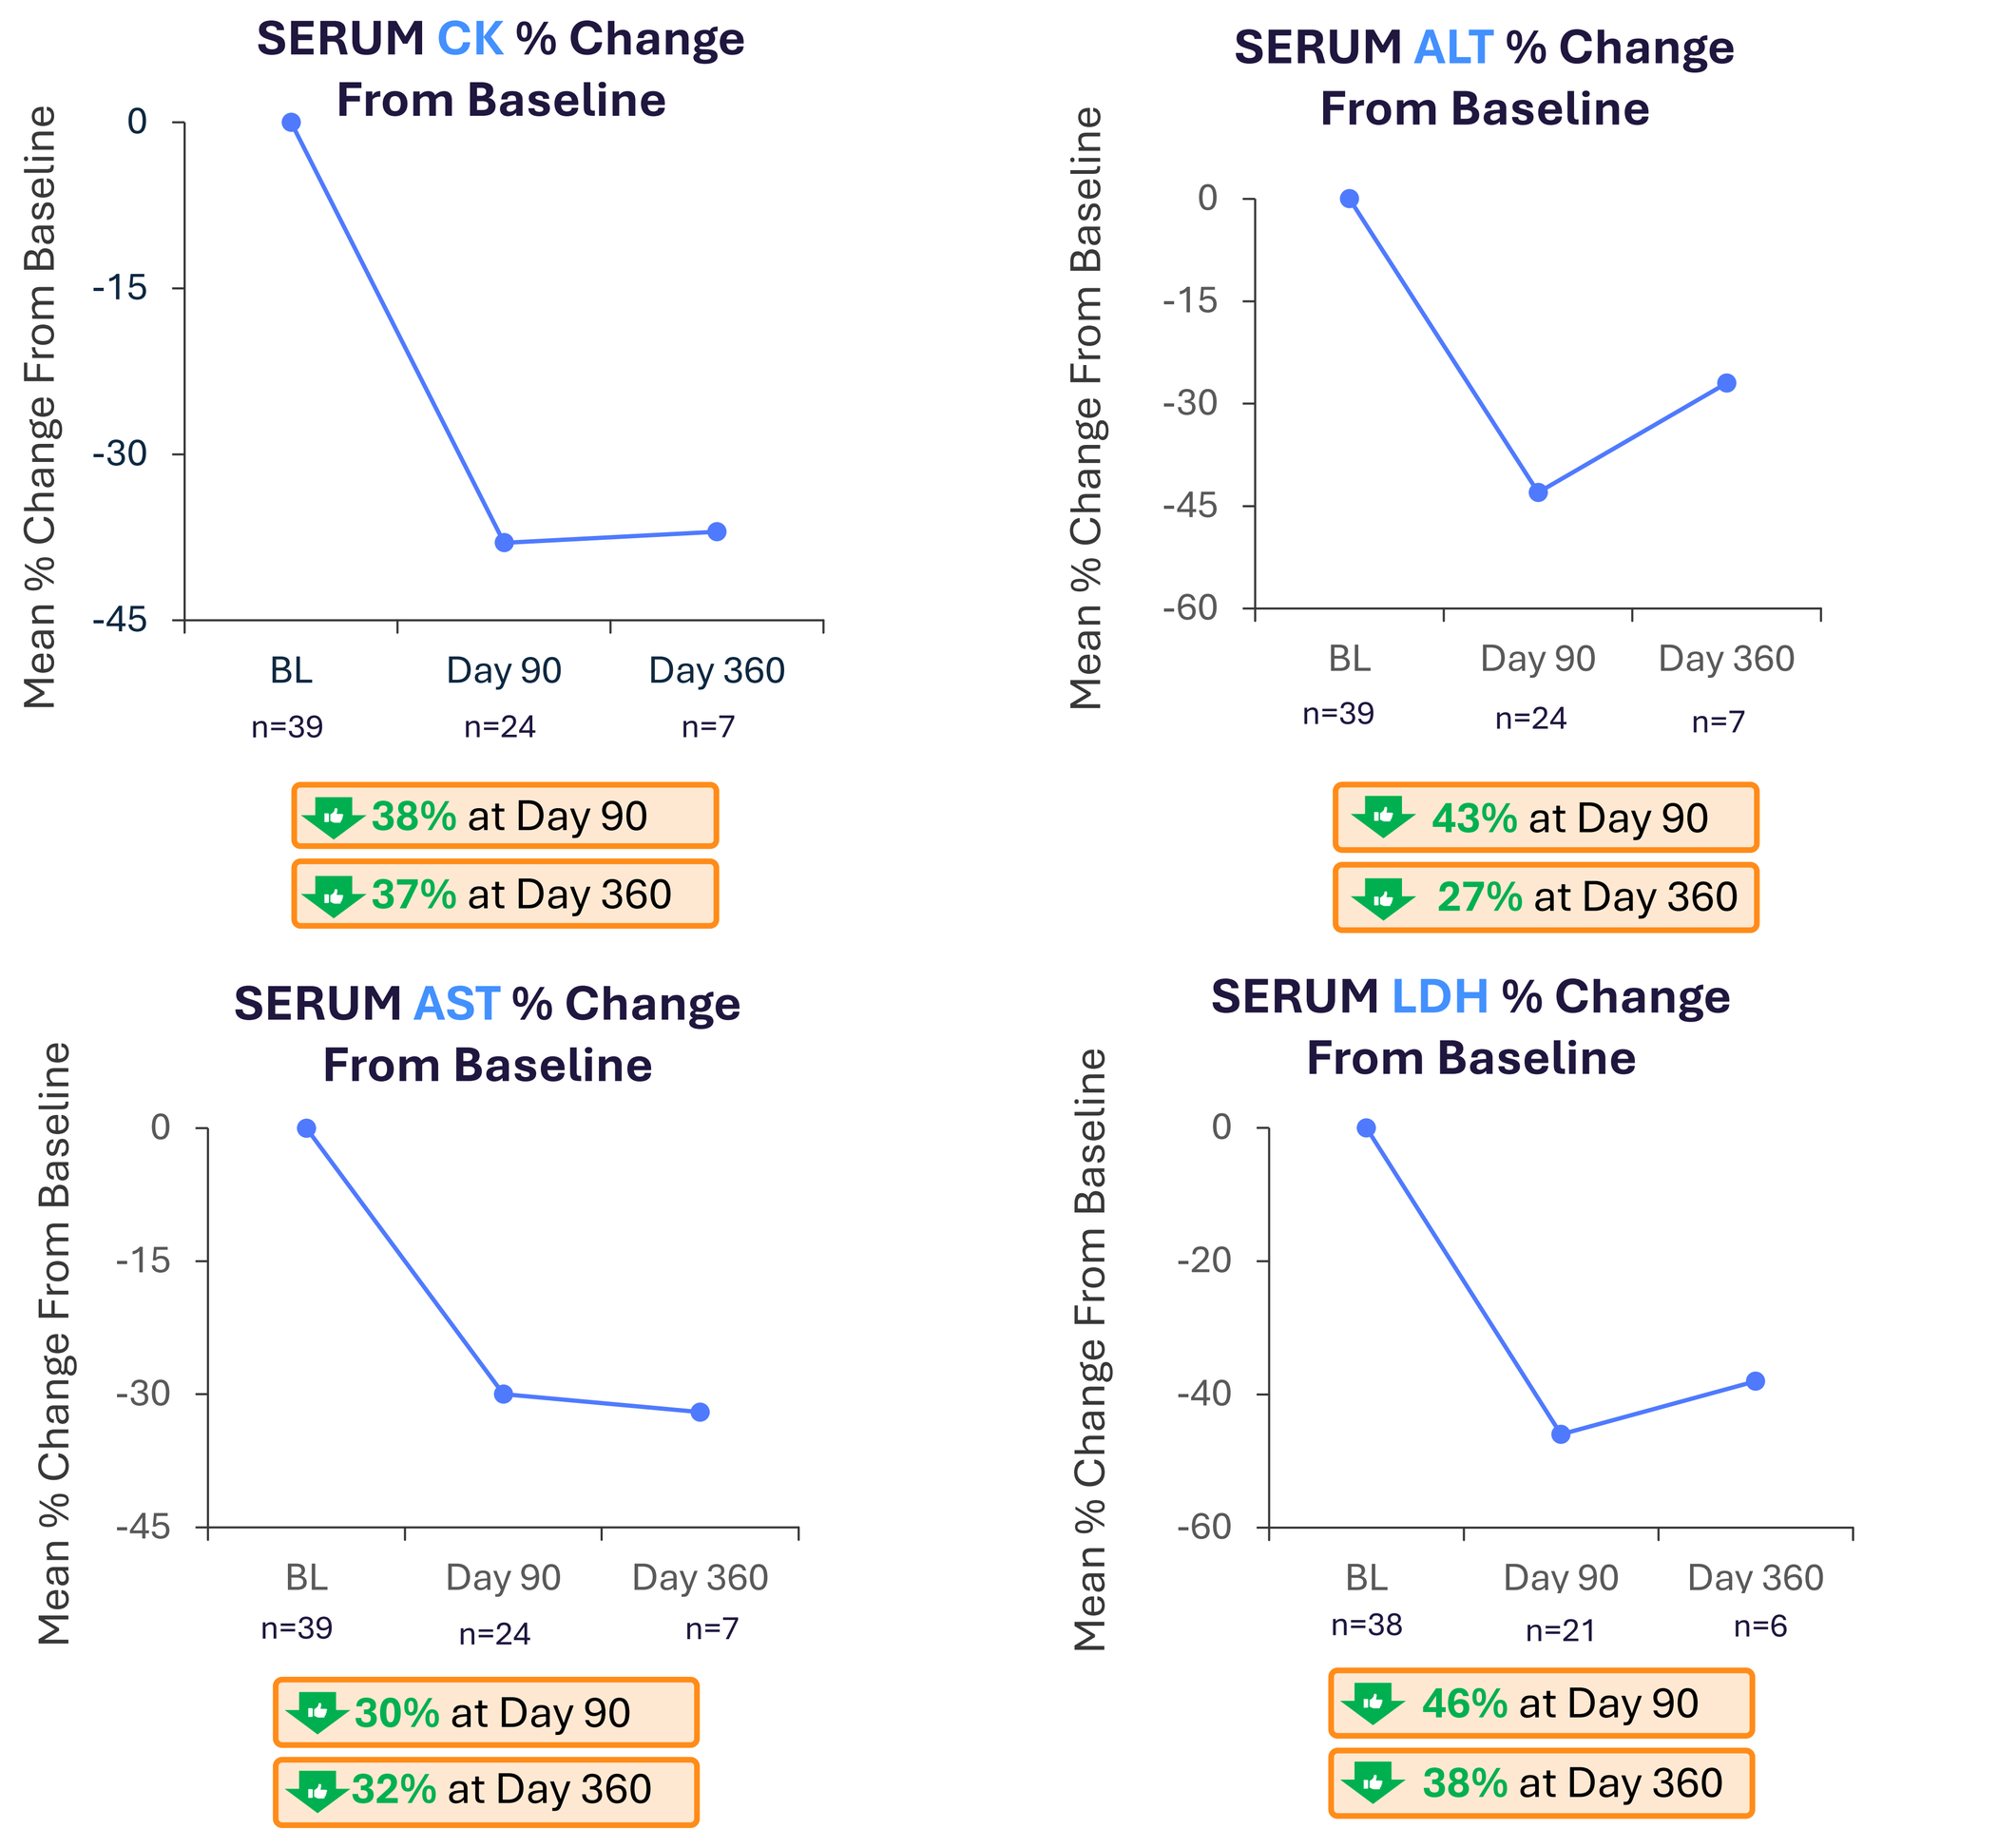
SERUM CK % Change From Baseline
Mean % Change From Baseline
0
-15
-30
-45
BL n=39
Day 90 n=24
Day 360 n=7
38% at Day 90
37% at Day 360
SERUM ALT % Change From Baseline
0
-15
-30
-45
-60
BL n=39
Day 90 n=24
Day 360 n=7
43% at Day 90
27% at Day 360
SERUM AST % Change From Baseline
0
-15
-30
-45
BL n=39
Day 90 n=24
Day 360 n=7
30% at Day 90
32% at Day 360
SERUM LDH % Change From Baseline
0
-20
-40
-60
BL n=38
Day 90 n=21
Day 360 n=6
46% at Day 90
38% at Day 360

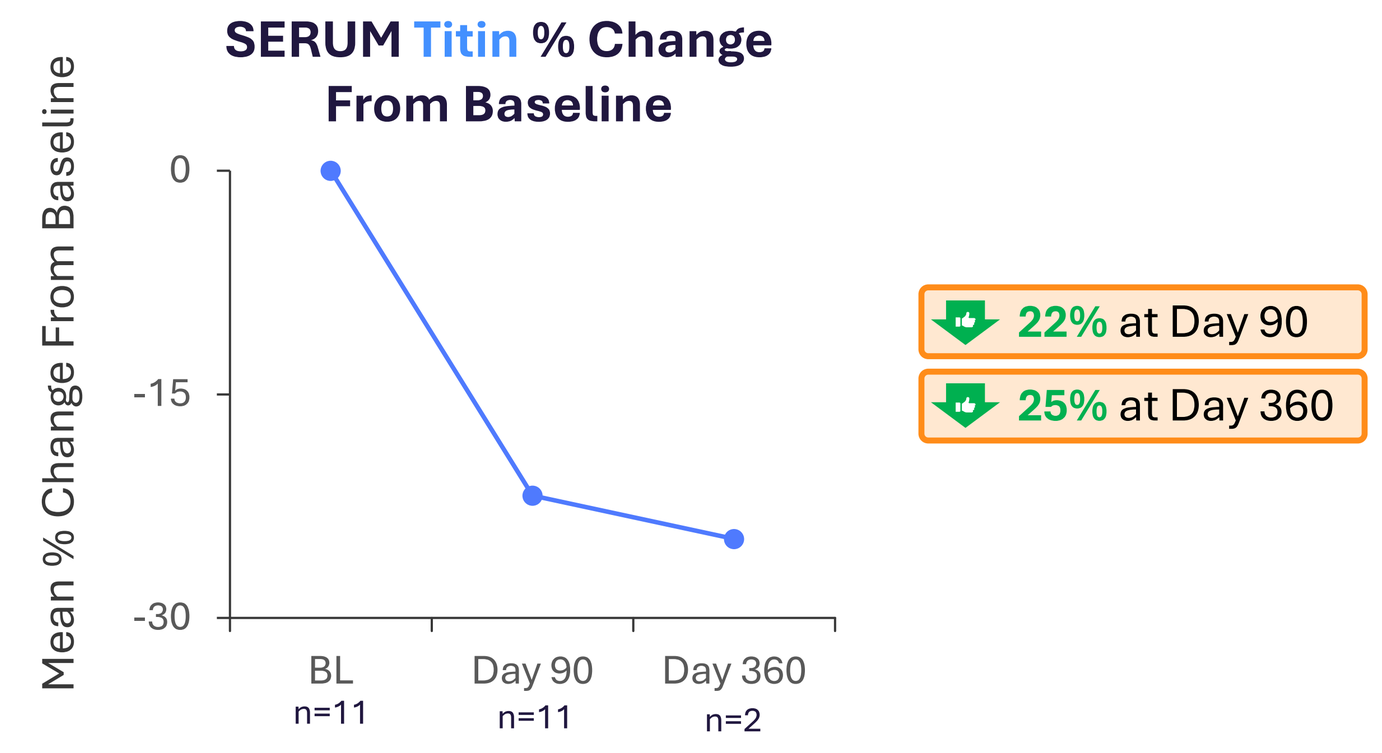
SERUM Titin % Change From Baseline
Mean % Change From Baseline
0
-15
-30
BL n=11
Day 90 n=11
Day 360 n=2
22% at Day 90
25% at Day 360

Figure 5. Improved muscle integrity may support slower disease progression and better long-term clinical outcomes 2-4. Certain data from a subset of participants were not available at the time of analysis.

STABLE TO IMPROVED CARDIAC FUNCTION OBSERVED AFTER SGT-003 TREATMENT

Figure 6. Observations of Cardiac Function and Cardiac Injury Markers Following SGT-003 Treatment[11]

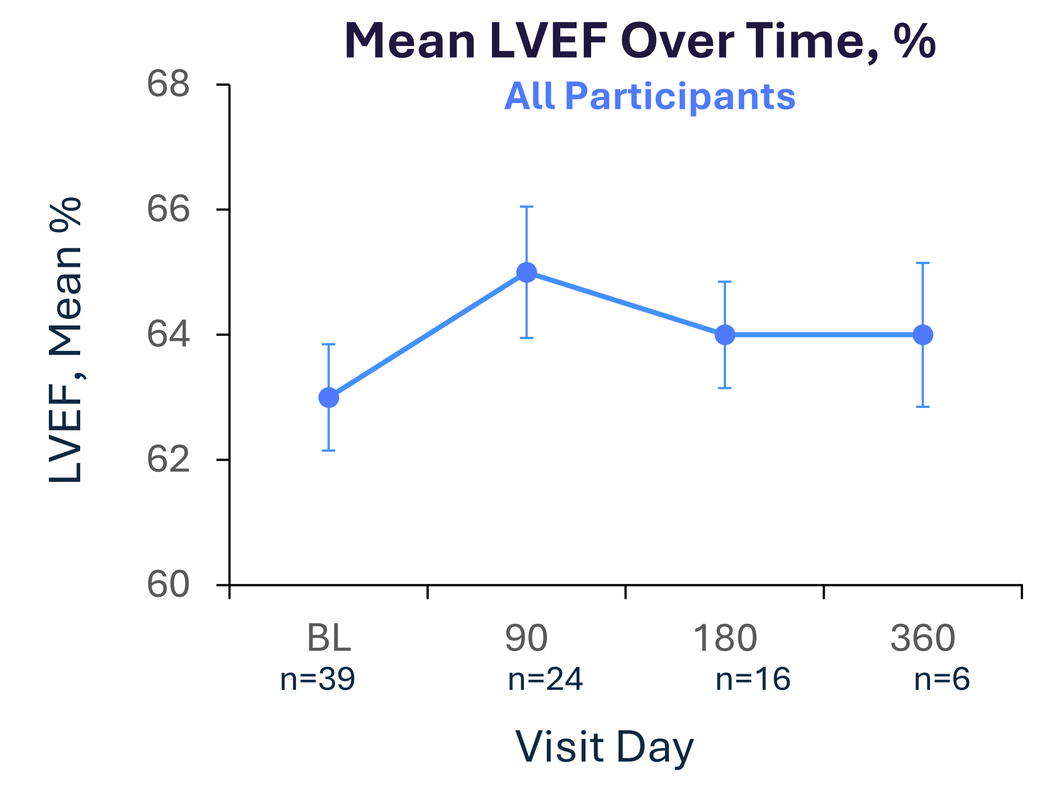
Mean LVEF Over Time, %
All Participants
LVEF, Mean %
68
66
64
62
60
BL n=39
90 n=24
180 n=16
360 n=6
Visit Day

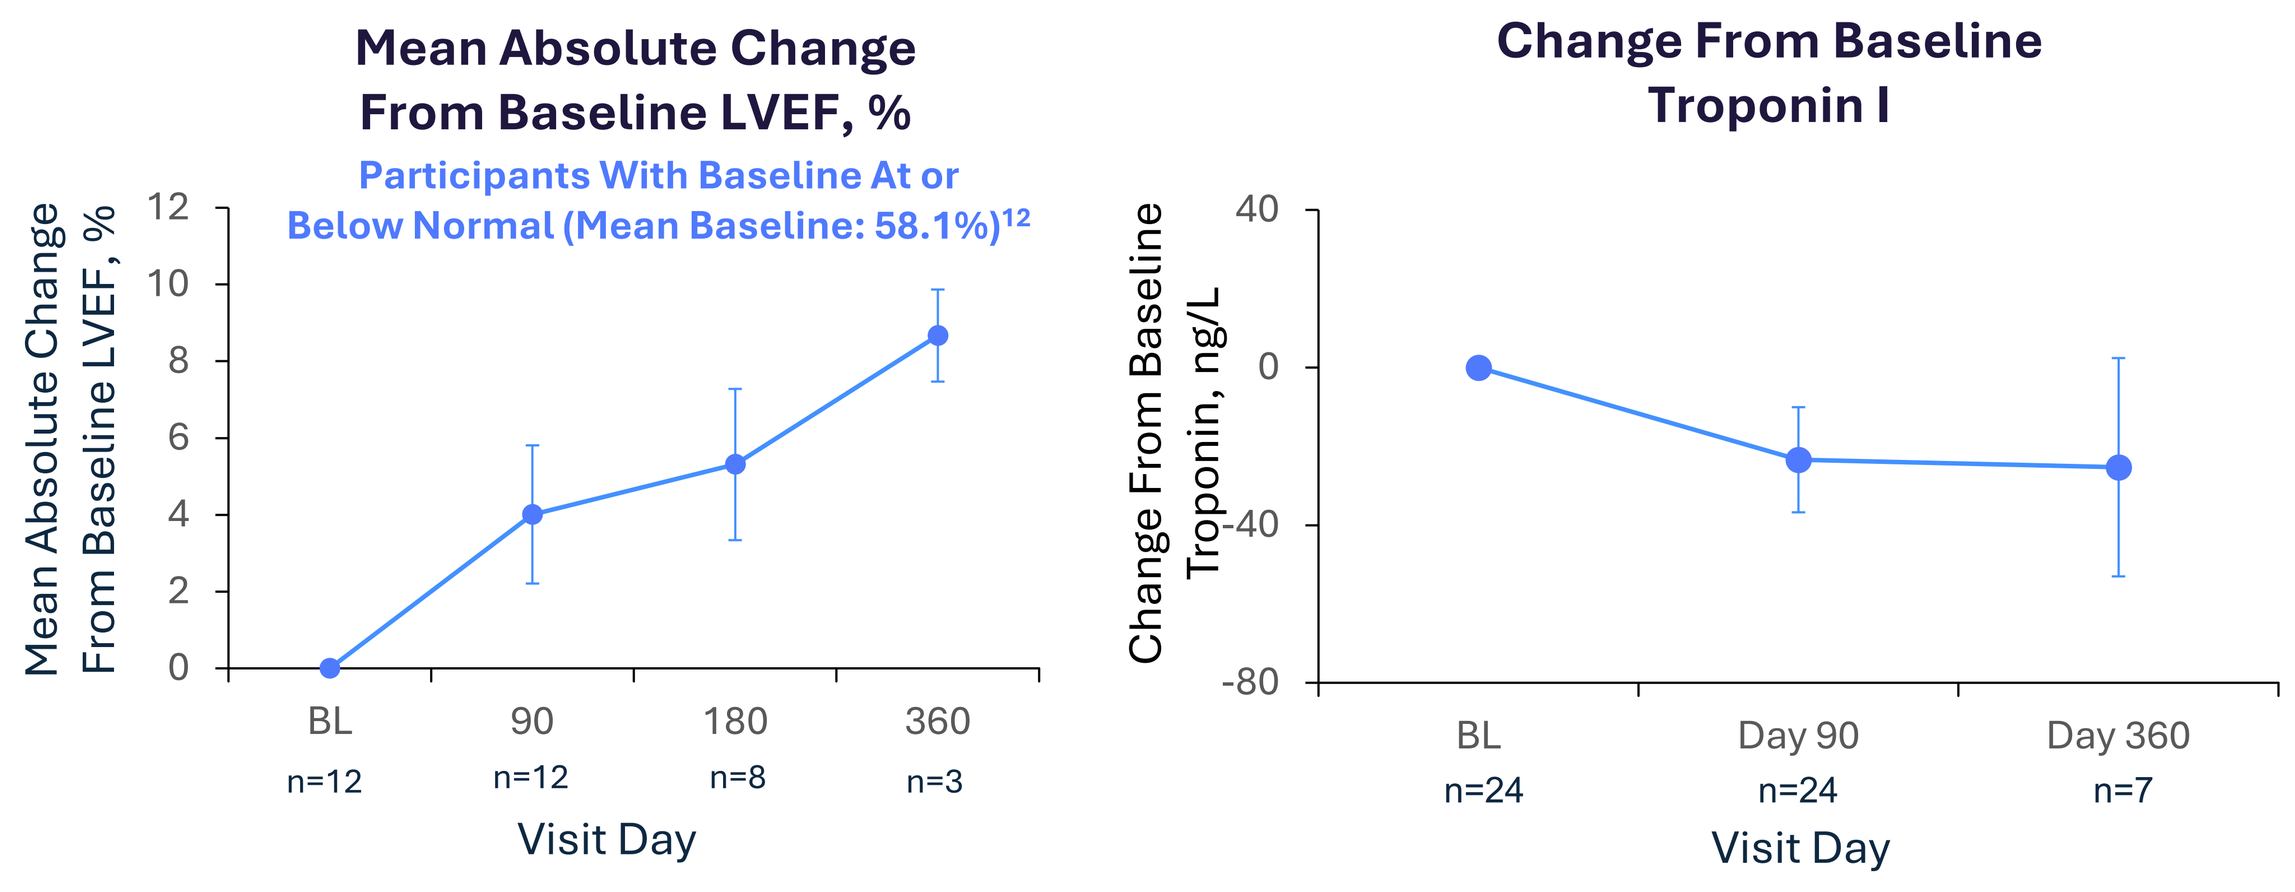
Mean Absolute Change From Baseline LVEF, %
Participants With Baseline At or Below Normal (Mean Baseline: 58.1%)12
Mean Absolute Change From Baseline LVEF, %
12
10
8
6
4
2
0
BL n=12
90 n=12
180 n=8
360 n=3
Visit Day
Change From Baseline Troponin I
Change From Baseline Troponin, ng/L
40
0
-40
-80
BL n=24
Day 90 n=24
Day 360 n=7
Visit Day

Figure 6. Observations of improved cardiac function are driven by participants with low-normal baseline LVEF[13]. Changes in troponin I levels were driven by participants with elevated troponin at baseline

CONCLUSIONS

- SGT-003 has been generally well tolerated in the 53 participants dosed in the INSPIRE DUCHENNE study as of August 4, 2026, using a steroid-only prophylactic immunomodulation regimen
- Robust transduction and microdystrophin expression have been observed in muscle biopsy samples collected at Day 90 and Day 360 after treatment with SGT-003
- Corresponding restoration of key elements of the DAPC has been observed in muscle biopsy samples
- Improvements in markers of muscle integrity have been evident as early as Day 90 and persist to at least Day 360 after treatment with SGT-003
- Stable-to-improved cardiac function has been observed post treatment with SGT-003 as measured by LVEF

Presented at the 30th Annual World Muscle Society, September 29-October 3, 2026, Hiroshima, Japan