Full-Length Transgene Quantification Utilizing NanoMosaic Tessie Technology

Isabella Pajevic, Peter Webster

Department of Analytical Sciences, Solid Biosciences Inc., Charlestown, MA, USA

SOLID BIOSCIENCES

Scan for Capsid Information

INTRODUCTION

- Product characterization is an important aspect of adeno-associated virus (AAV)-based gene therapy development, where accurate quantification of full-length transgenes and capsid titers is essential for maintaining consistent quality, safety, and efficacy.
- Currently, the AAV field utilizes several approaches to quantify product integrity, including both molecular and biophysical methods.
- Combining data obtained from multiple systems compounds assay variability resulting in decreased accuracy, thereby lessening the value of the obtained results.

METHODS & MATERIALS: TESSIE PLATFORM

- To measure full-length transgene and empty/full ratio, improvements were made to the primer sequences for use with our custom transgene using the Tessie platform. The Tessie nanoneedle platform enables measurement of AAV transgenes and AAV capsids in one platform. The Tessie nano-plate nanoneedles are densely integrated on a silicon chip and are configured into 96- or 384-well formats for high-throughput workflows.
- We report the development of a viral genome assay utilizing two affinity-tagged oligonucleotide probes to selectively bind specific regions of the transgene. Through limited primer extension, these probes incorporate into a double-stranded deoxyribonucleic acid (ds DNA) product, enabling differentiation between full-length and truncated transgenes. Full-length genomes were detected by probes positioned at opposite ends of the therapeutic region. The resulting labeled DNA was quantified with high sensitivity using Tessie nanoneedle sensors having more than 20,000 nanoneedles in each well, resulting in an accurate and scalable method for assessing AAV genome integrity.

Figure 1. Graphic Display of NanoMosaic Tessie Nano-plate Format

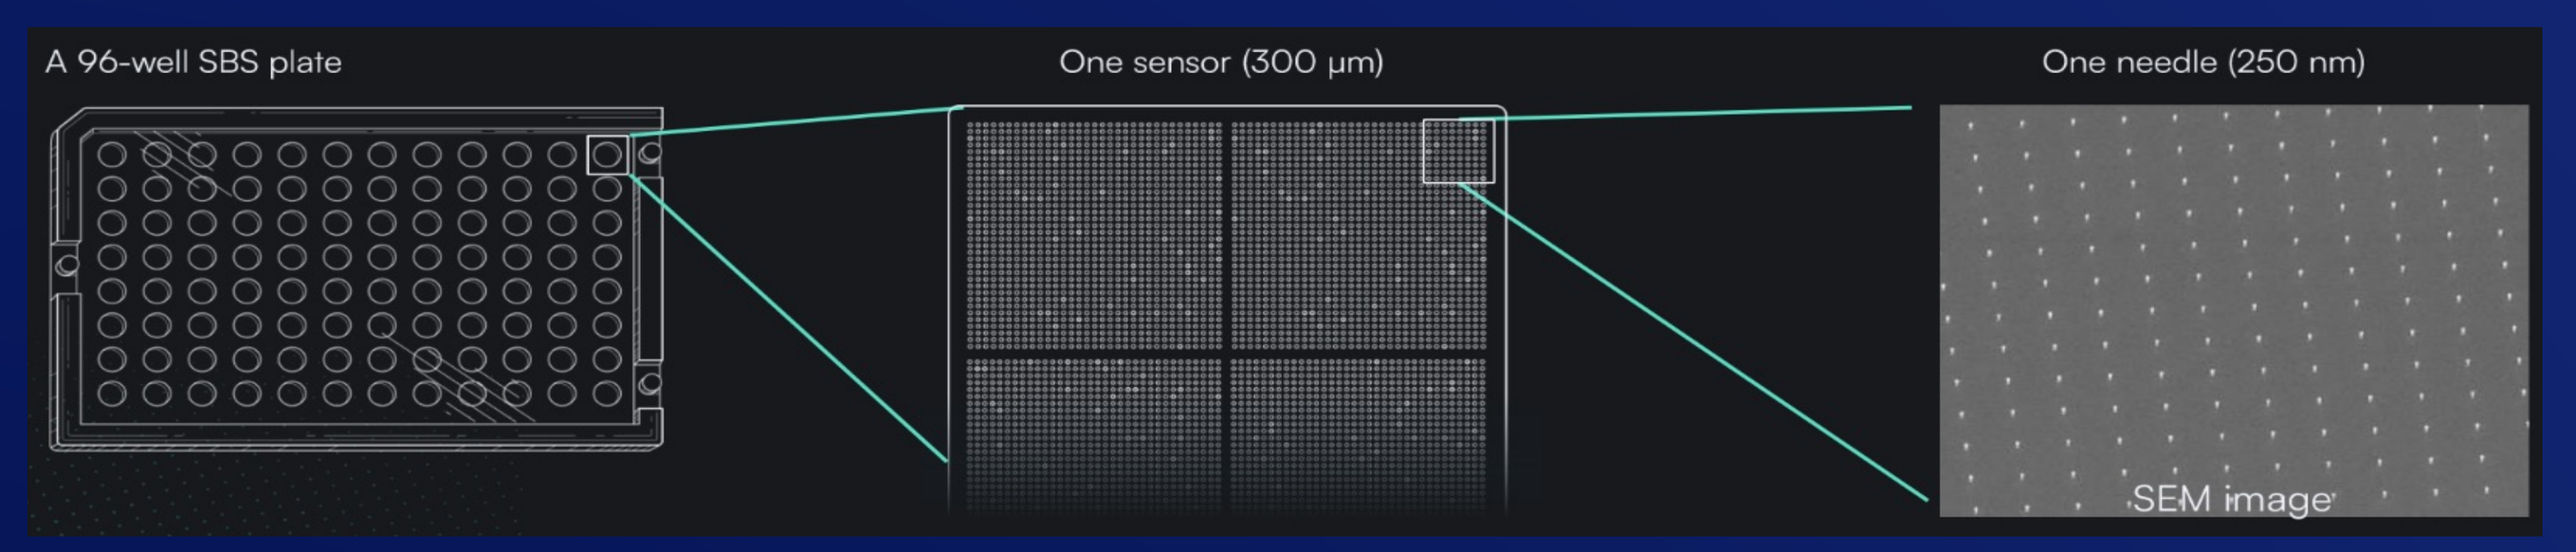
A 96-well SBS plate
One sensor (300 µm)
One needle (250 nm)
SEM image

Figure 2. Graphic Display of Simultaneous Binding and Quantification Capabilities on the Tessie Nano-plate Nanoneedle

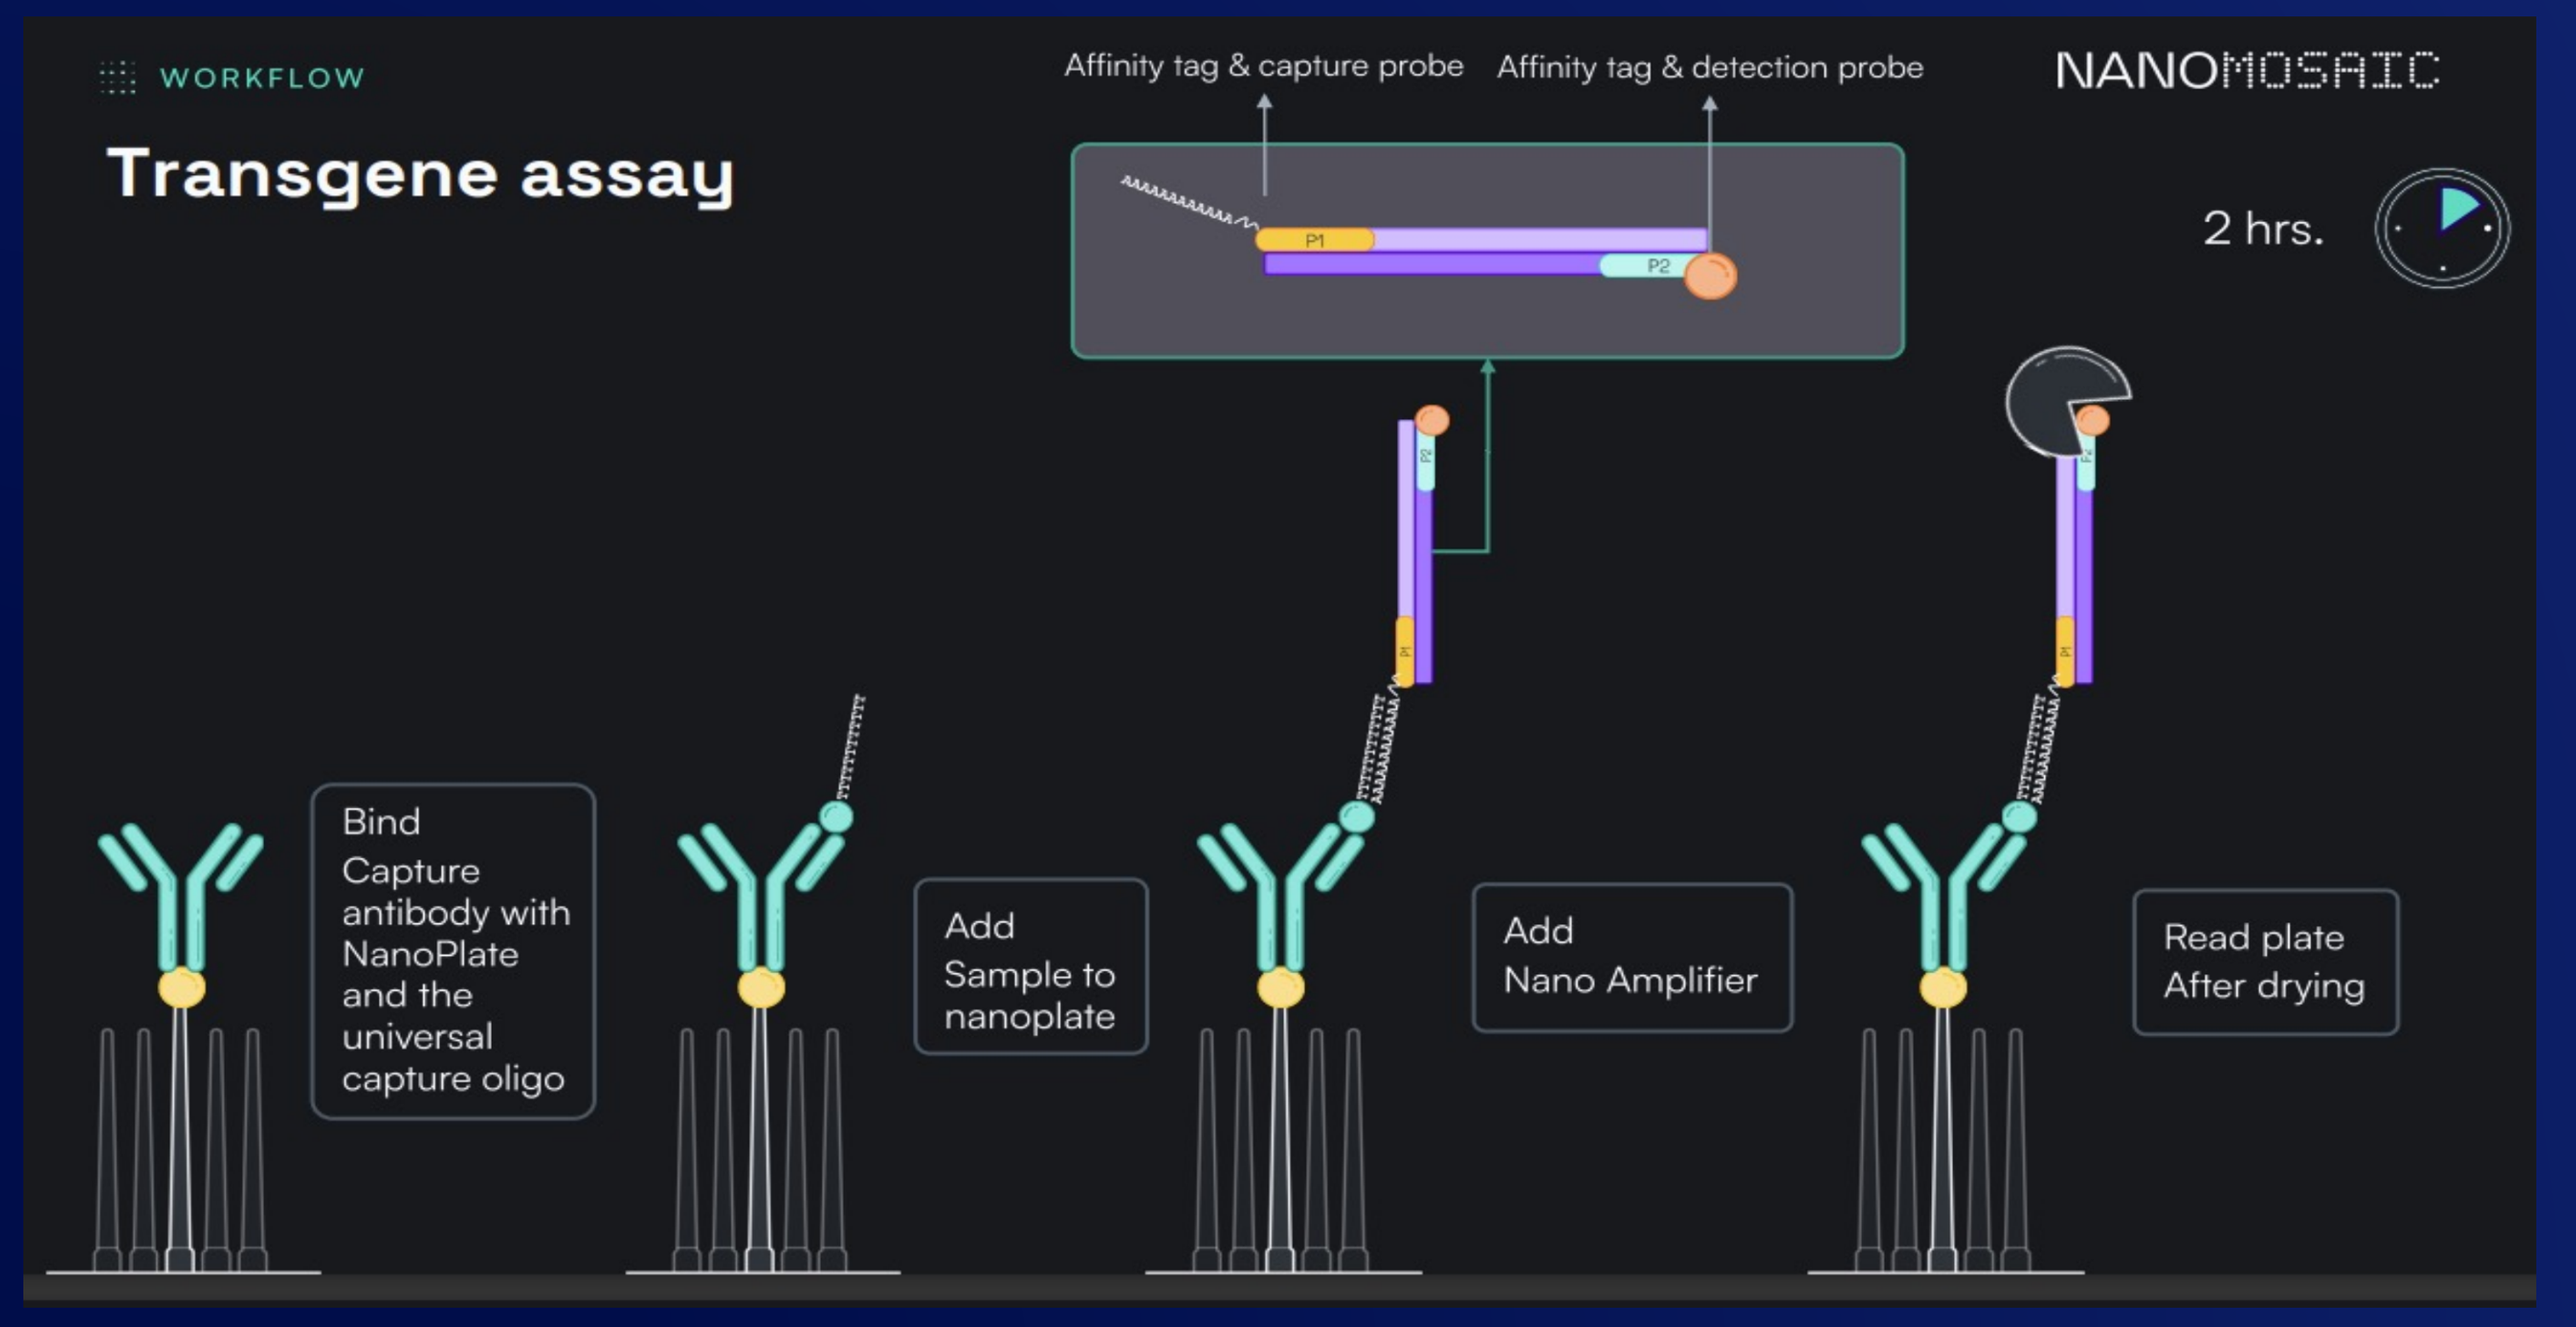
WORKFLOW
Transgene assay
Affinity tag & capture probe
Affinity tag & detection probe
NANOMOSAIC
2 hrs.
Bind Capture antibody with NanoPlate and the universal capture oligo
Add Sample to nanoplate
Add Nano Amplifier
Read plate After drying

DESIGN

STANDARD CURVE OPTIMIZATION

- Optimizing the curve is important for accurate quantification of full-length transgenes with a standard curve-based assay. The full-length transgene assay utilizes a standard curve for relative quantification of a test article. Due to the relative nature of the assay, optimizing and characterizing the standard is critical for assay performance.
- Solid utilized a linearized plasmid of known full-length percentage as the reference standard and optimized the standard curve for range, linearity, and accuracy. Identifying the linear range within that curve is important for determining the range of titers the assay can reliably measure. To optimize this, different amplification cycles were tested to find the point where sample signals fall within the linear portion of the curve, avoiding oversaturation or excessive dilution.

Figure 3. Standard Curve Optimization

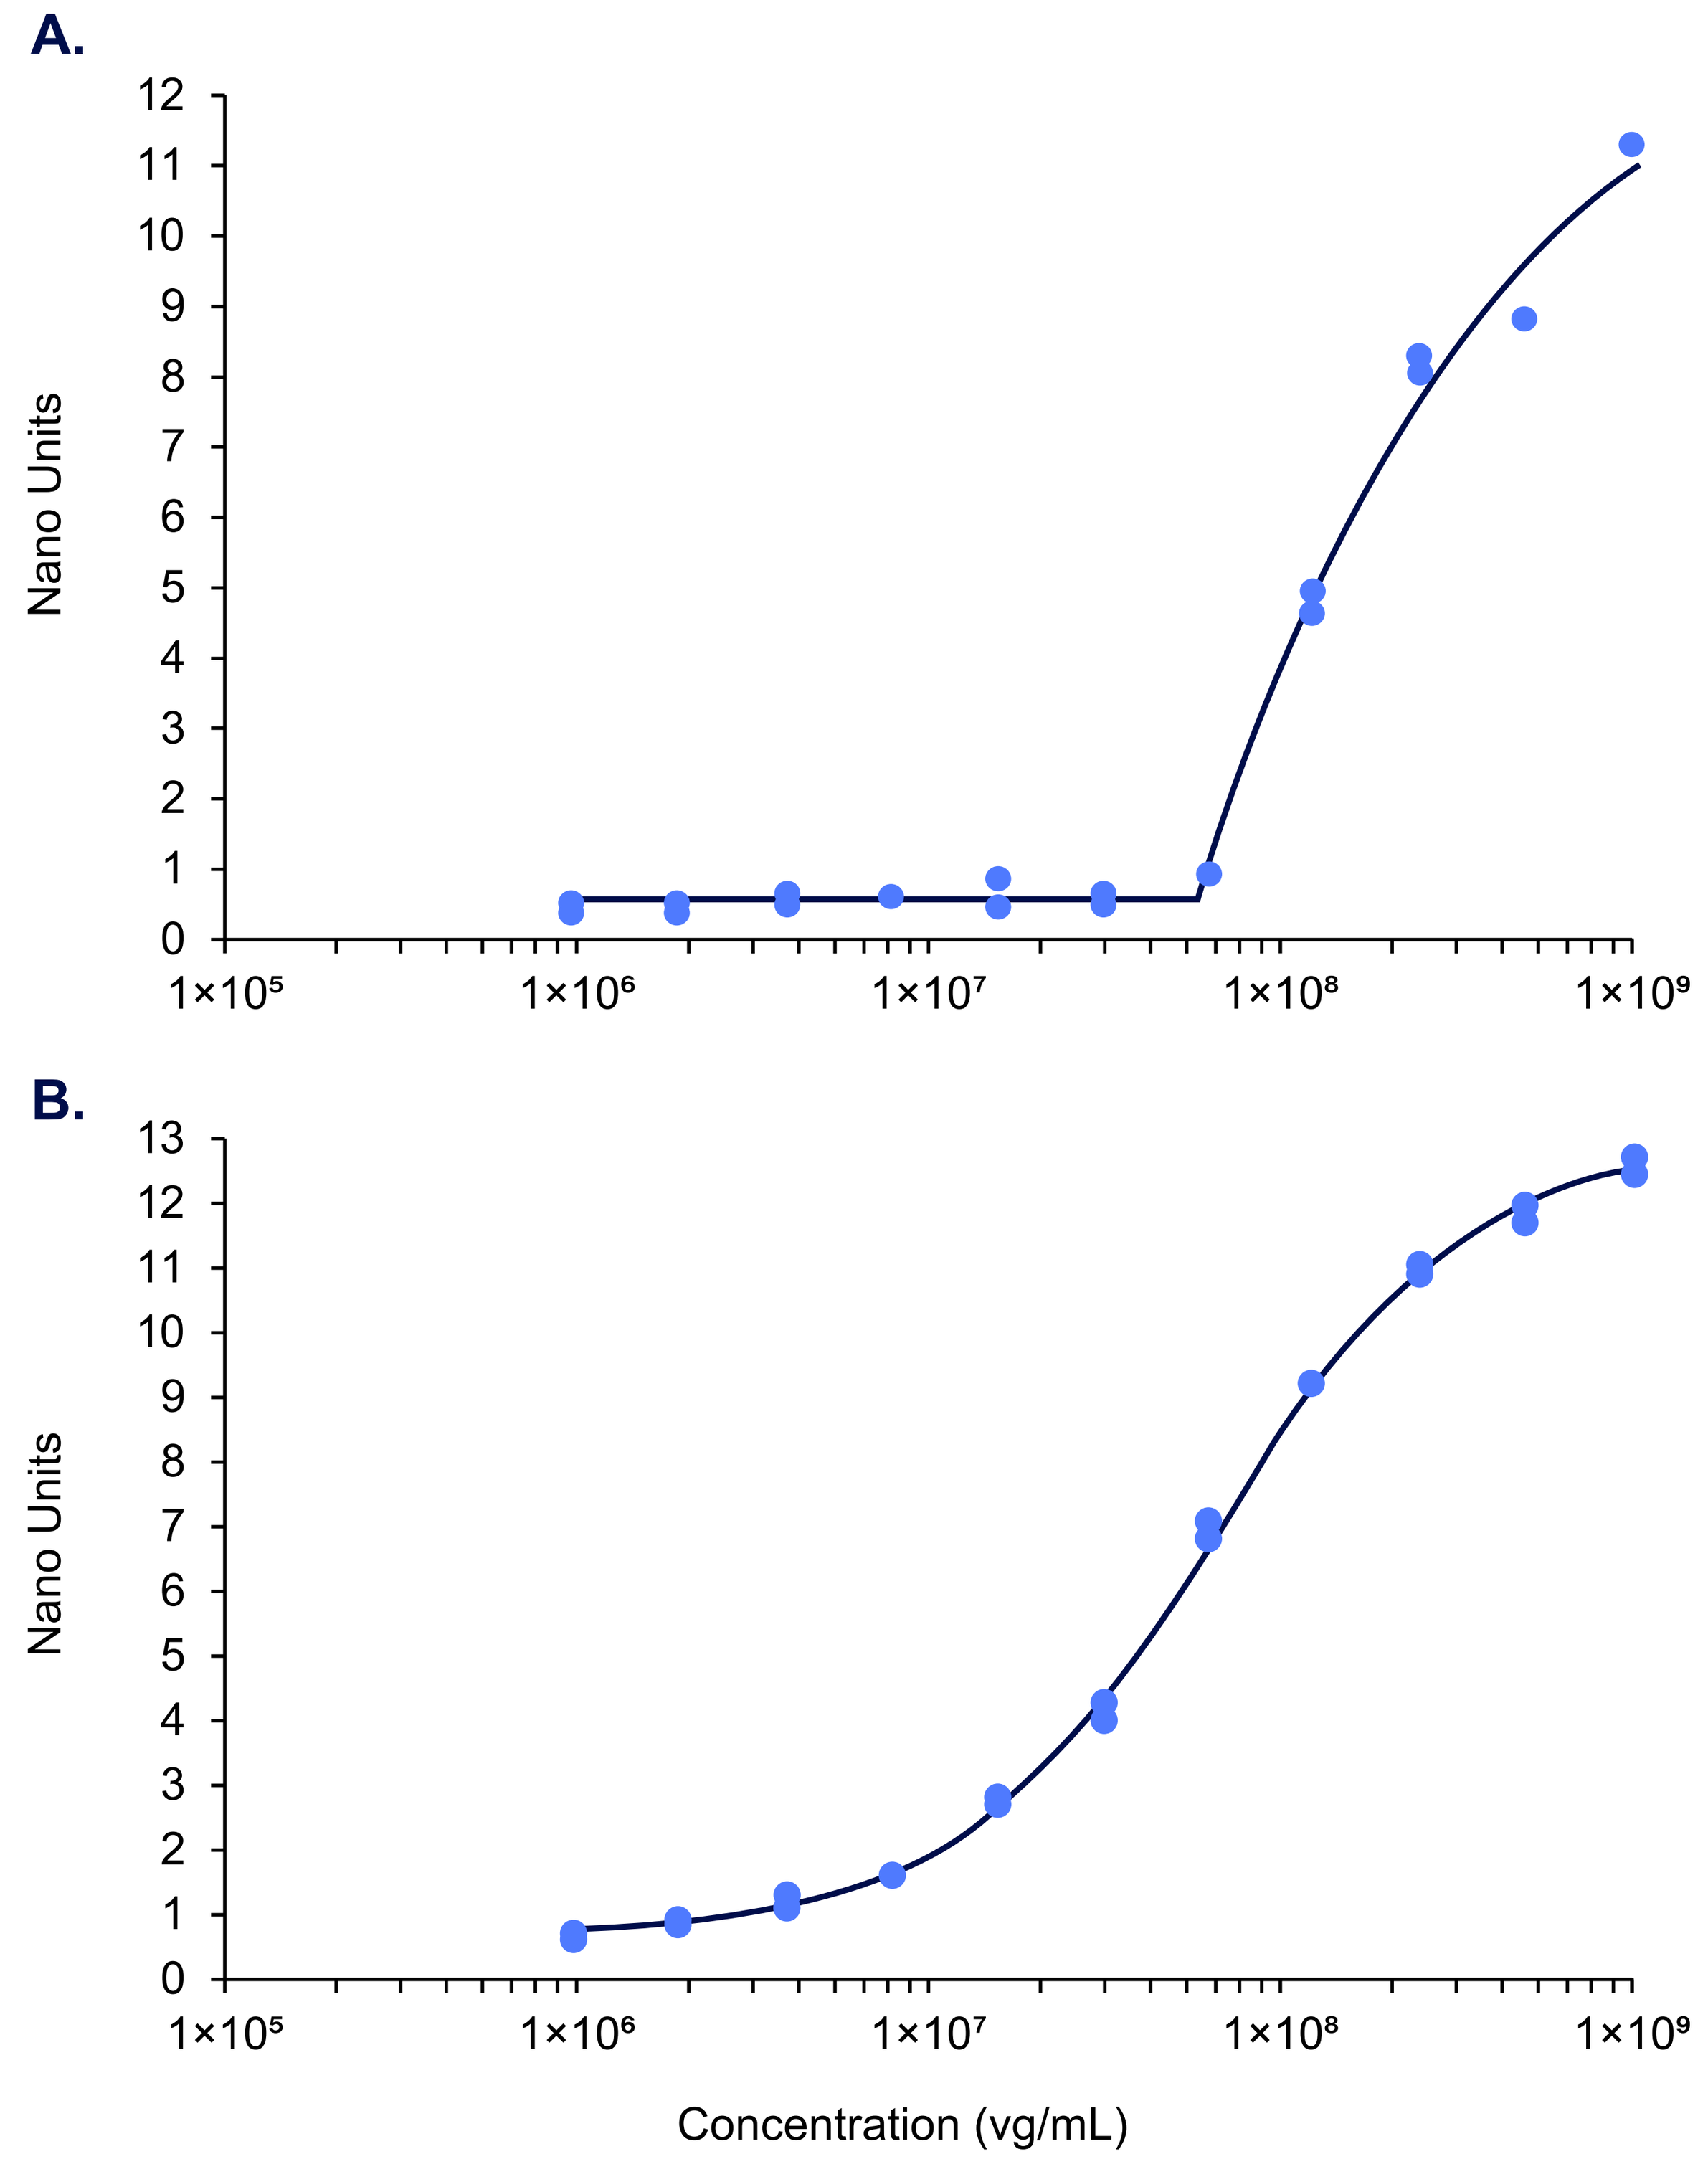
A.
B.
Nano Units
Nano Units
1×10⁵
1×10⁶
1×10⁷
1×10⁸
1×10⁹
Concentration (vg/mL)

Standard curve graphs amplified 14 cycles. **Curve A** displays oversaturation with only a few data points falling within the linear range, limiting its usefulness for accurate quantification. **Curve B** displays an ideal shape with a well-defined linear range, allowing more reliable measurement across different sample concentrations. Probe concentration was adjusted in **Curve B** to improve the results, which enhanced signal clarity and expanded the usable dynamic range.

TRANSGENE QUANTIFICATION

FULL-LENGTH TRANSGENE QUANTIFICATION

- Measuring the full-length genome provides a more accurate indication of the vector's ability to produce full-length mRNA and functional protein. Traditional ddPCR methods that target internal regions may detect partial or fragmented genomes, leading to an overestimation of intact vector content. By focusing on full-length sequences, this method may better correlate with protein expression levels and overall vector potency. It offers a more precise assessment of genome integrity compared to conventional approaches.

Figure 4. Full-Length Transgene Results vs Traditional ddPCR Results

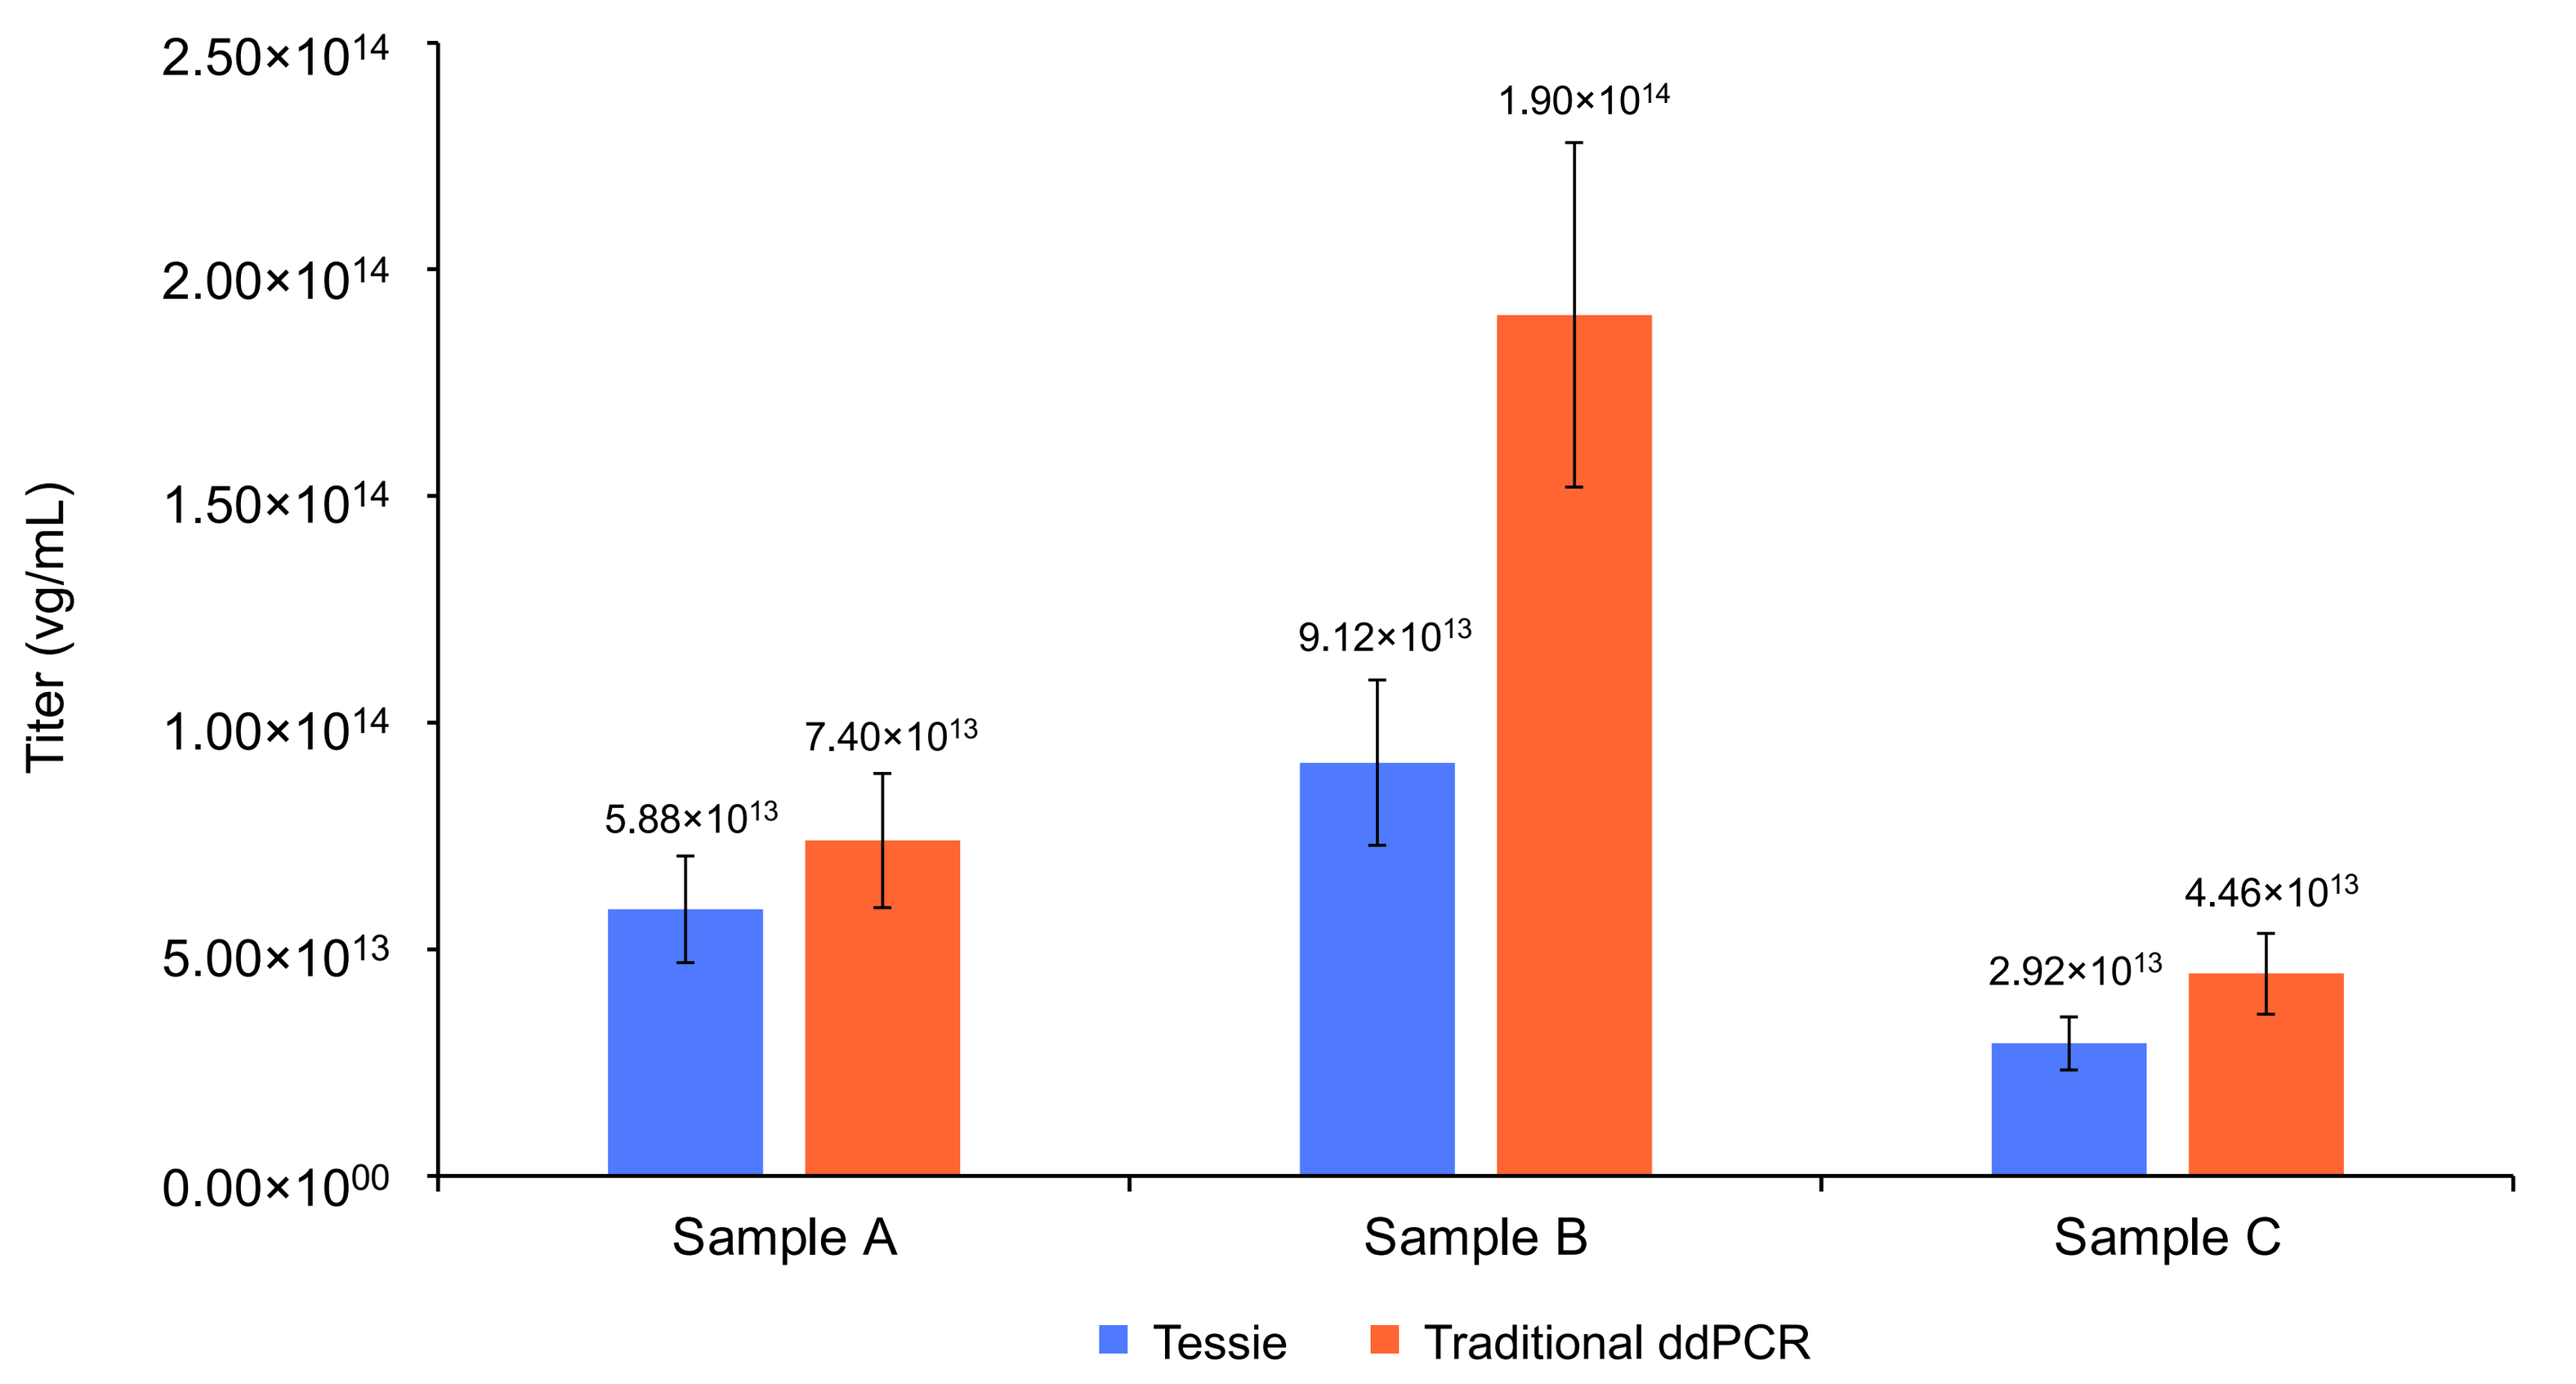
Titer (vg/mL)
2.50×10¹⁴
2.00×10¹⁴
1.50×10¹⁴
1.00×10¹⁴
5.00×10¹³
0.00×10⁰⁰
5.88×10¹³
7.40×10¹³
9.12×10¹³
1.90×10¹⁴
2.92×10¹³
4.46×10¹³
Sample A
Sample B
Sample C
Tessie
Traditional ddPCR

Full-length Tessie transgene results compared to traditional ddPCR transgene results across three samples. The results showed consistent lower full-length transgene titer compared to traditional ddPCR, demonstrating that traditional ddPCR may overestimate the viral genome titer. The large difference seen in Sample B, as compared to Samples A and C, may indicate fragmented or truncated transgene being detected in Sample B by traditional ddPCR.

DESIGNING PRIMERS FOR THE TESSIE METHOD

- To utilize this method for full-length transgene quantification, affinity-tagged oligonucleotide probes were designed to target the 5' and 3' ends of the transgene. Unlike traditional ddPCR primer design, which typically targets a short internal region of the transgene, this method requires simultaneous binding at both the 5' and 3' ends to ensure that only intact, full-length sequences are amplified. This reduces the likelihood of detecting partial genomes, offering greater specificity for assessing transgene integrity.

Figure 5. An Example Genome with 5' and 3' Primer Targets, Along with Traditional ddPCR Binding Site

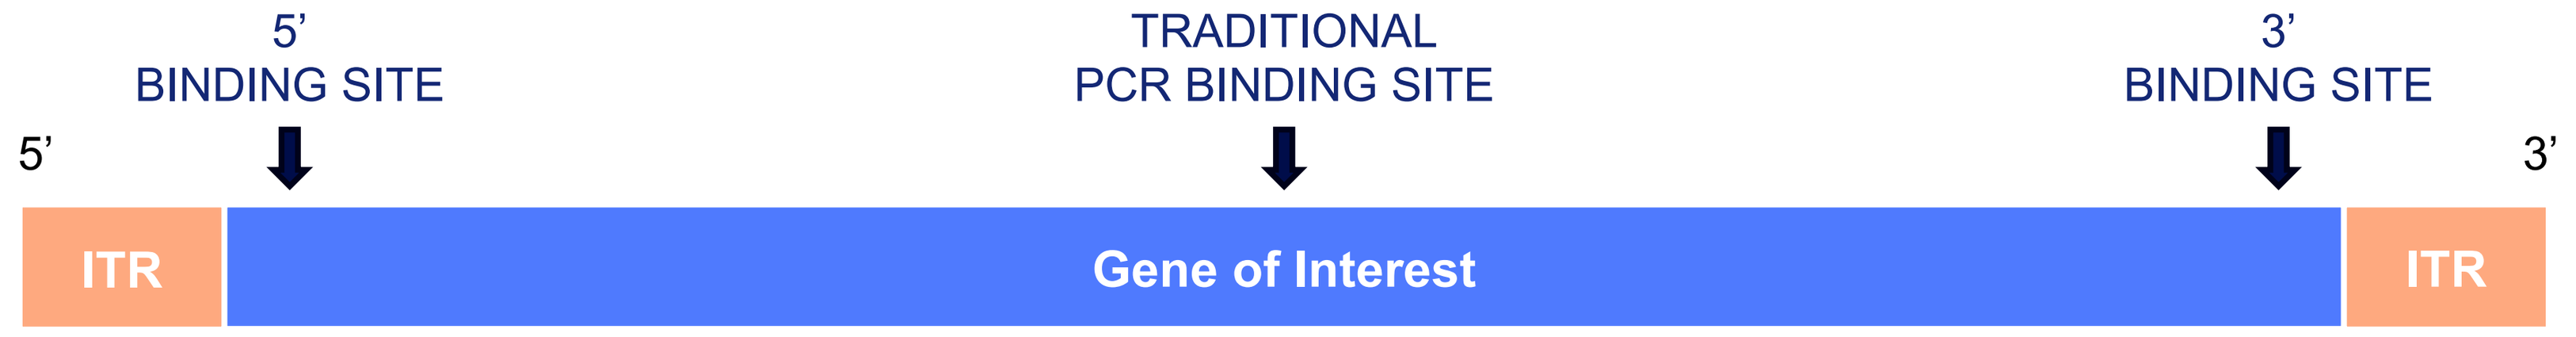
5'
BINDING SITE
TRADITIONAL
PCR BINDING SITE
3'
BINDING SITE
5'
3'
ITR
Gene of Interest
ITR

By measuring from the 5' to 3' end, this platform can quantify full-length transgene. As seen in **Figure 4**, full-length transgene titers are consistently lower than those measured by traditional ddPCR across multiple samples. This shows that traditional ddPCR may overestimate genome content by detecting partial or fragmented sequences, while the full-length approach offers a more complete measure of genome integrity.

EMPTY/FULL RATIO

ORTHOGONAL METHODS

- Orthogonal methods for measuring empty/full capsid ratios include Analytical Ultracentrifugation (AUC) and CryoTEM. While AUC and CryoTEM provide valuable structural information and can distinguish empty from full particles, they cannot confirm whether the encapsidated genome is complete or specific to the intended transgene. Both techniques lack the ability to characterize the DNA identity, unlike the Tessie transgene/capsid titer assessment, which offers a more comprehensive assessment of vector quality.

CALCULATION OF EMPTY/FULL RATIO

- The empty/full ratio is an important quality attribute in gene therapy, as it reflects the proportion of viral particles that contain a functional, full-length genome versus empty capsids. There are several methods to calculate this ratio, including using the full-length transgene generated from the Tessie or AUC data.

Figure 6. Full Percentage

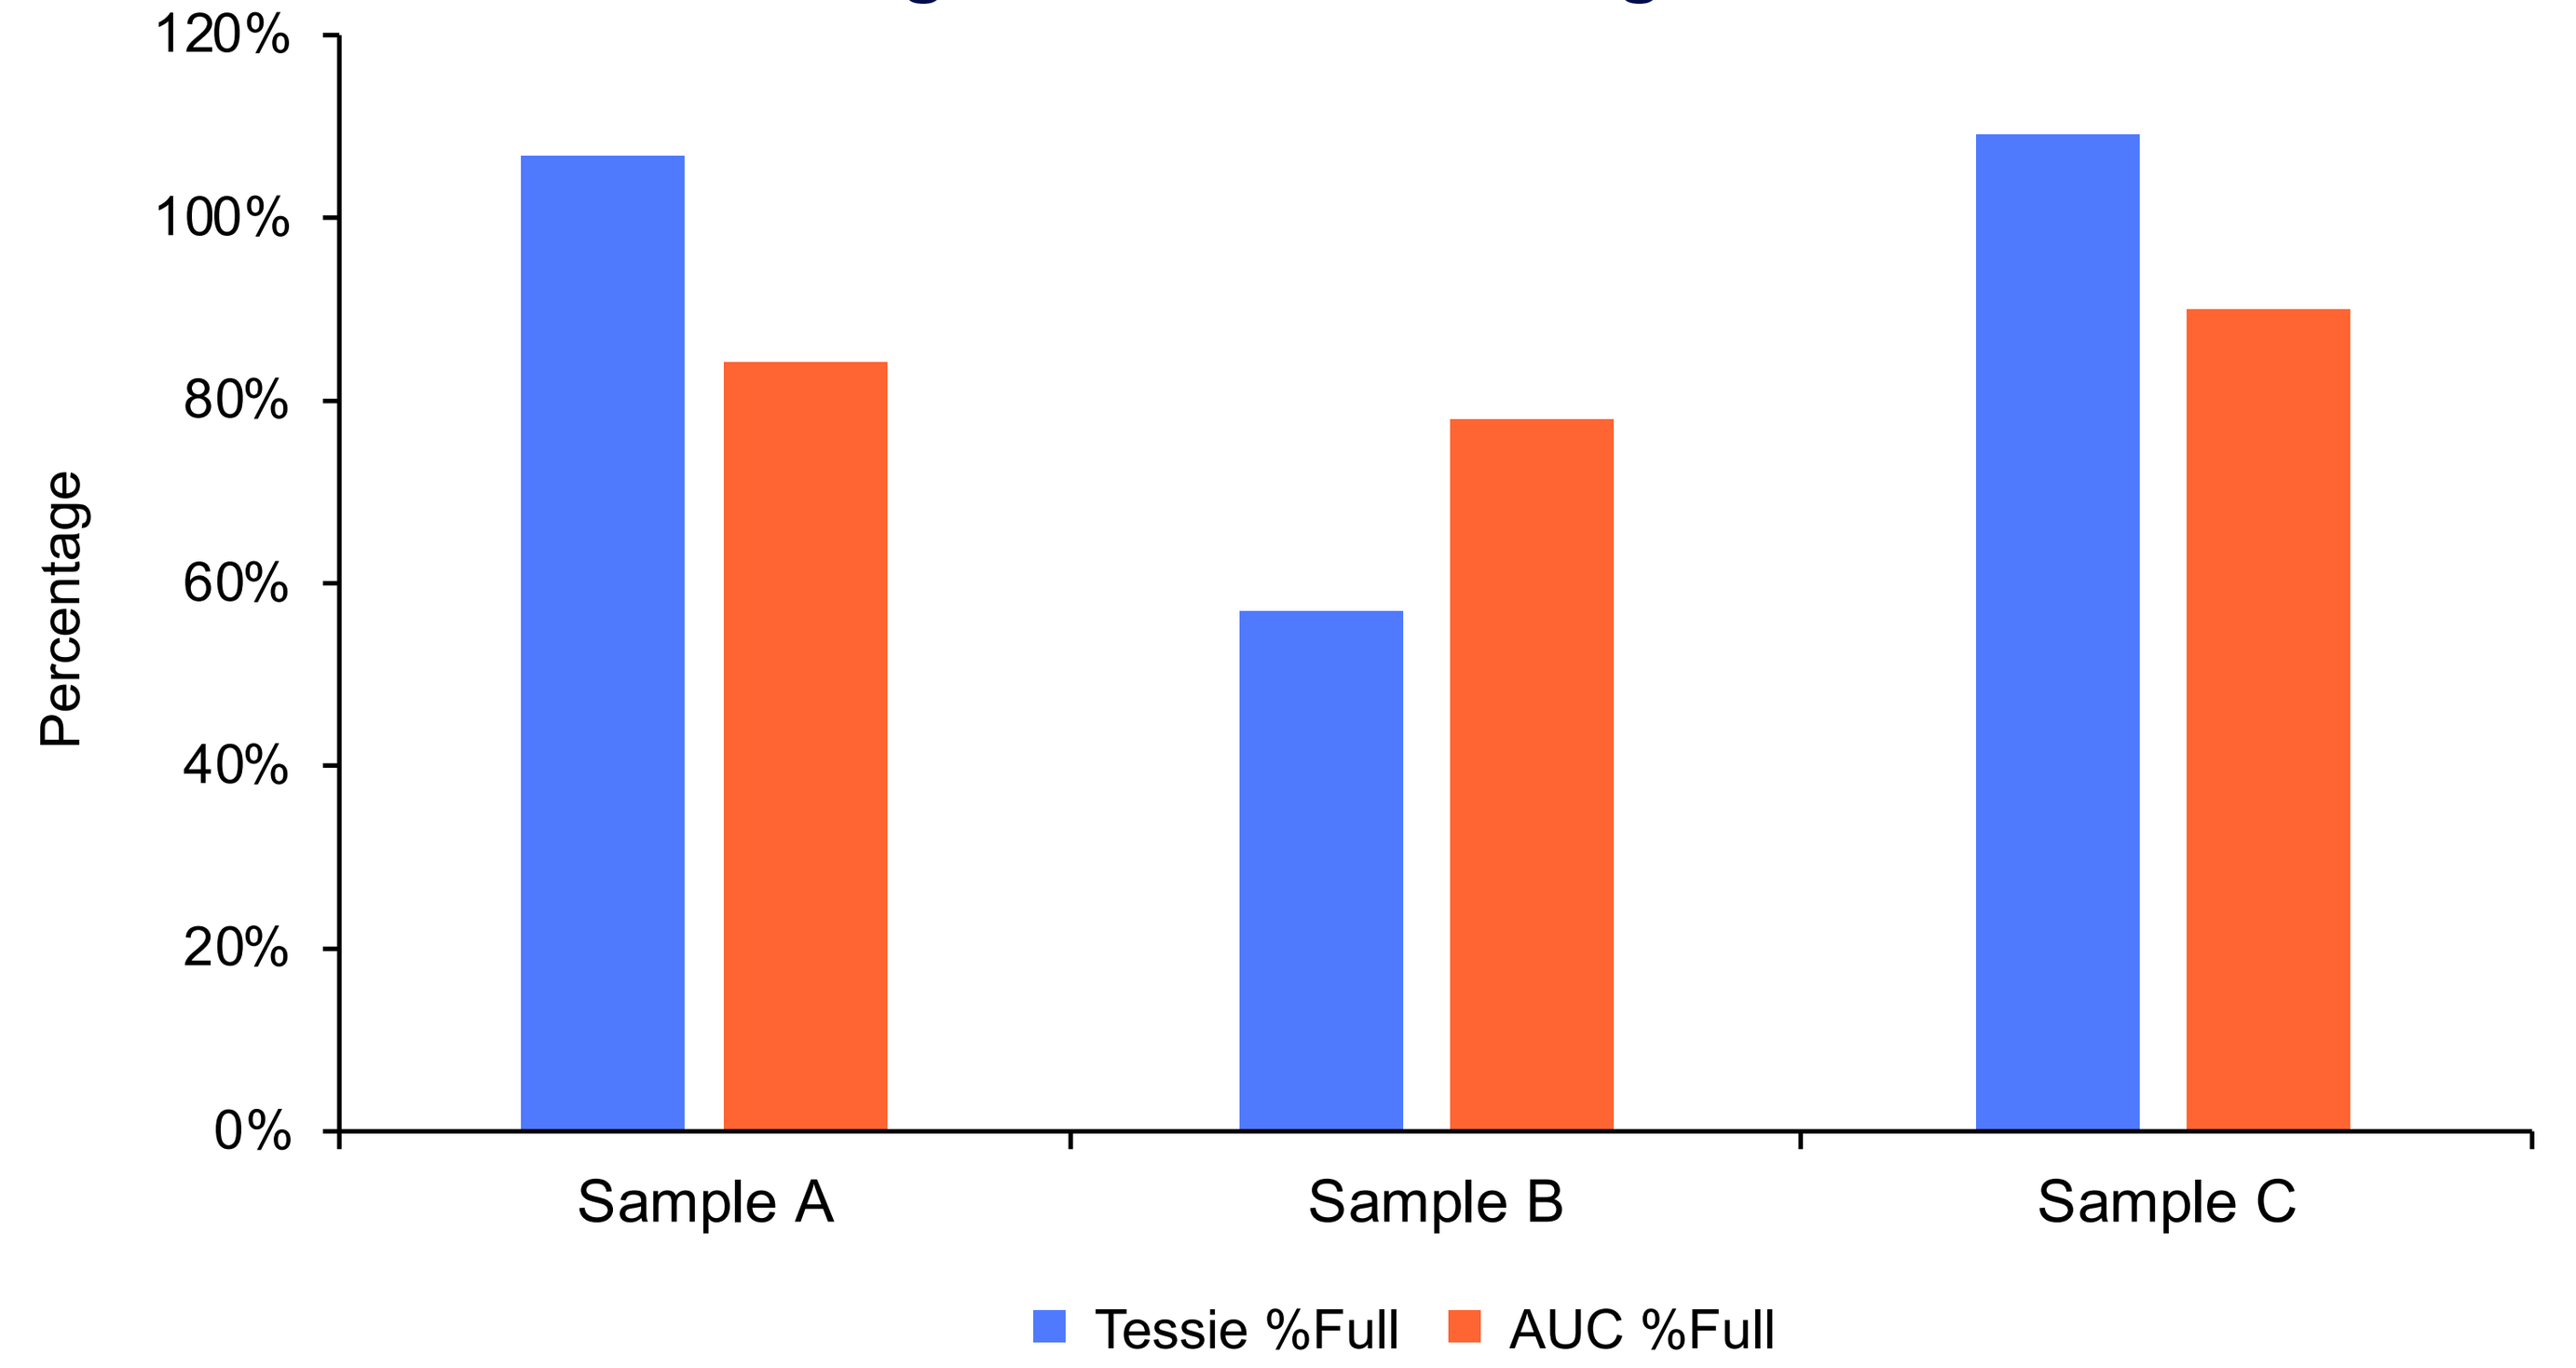
Percentage
120%
100%
80%
60%
40%
20%
0%
Sample A
Sample B
Sample C
Tessie %Full
AUC %Full

Comparison of percent full capsids for three samples as determined by Tessie and AUC. Samples A and C have a higher Tessie-based percent full, while Sample B shows a higher AUC-based percent full, suggesting both methods have some variability. The full percentage appears to exceed 100% for Samples A and C, which can occur when capsid and transgene titers are very similar and can be due to assay variability.

CONCLUSIONS & NEXT STEPS

- As the AAV and gene therapy field has matured, it has become clear that quantifying full-length genomes is important for thorough product characterization and understanding overall product strength. The initial development of a full-length transgene assay using the Tessie platform has shown that it is a robust and viable method for accurately detecting intact transgenes in AAV products.
- This technique enables more informed process development by providing early insights, such as identifying constructs with lower truncation propensity or manufacturing processes that better preserve transgene integrity. Ultimately, incorporating full-length genome quantification supports more consistent and higher-quality AAV production.

Presented at the ASGCT 28th Annual Meeting, May 13-17, 2025, New Orleans, LA, USA.