Capillary Isoelectric Focusing (cIEF) Platform for Characterization of Charge Variants of Adeno-Associated Virus (AAV) Capsids and Impact on Their Transduction Efficiency

Brandon Hoyle, Jonathan Hill

Department of Analytical Sciences, Solid Biosciences Inc., Charlestown, MA, USA

SOLID BIOSCIENCES

Scan for Capsid Information

INTRODUCTION

- Capillary isoelectric focusing (cIEF) has been growing as a standard method to evaluate charge heterogeneity of adeno-associated virus (AAV) vectors. However, cIEF methods may only be suitable for one or a couple of AAV serotypes, thus limiting characterization over a wide range of other AAVs.
- The present platform cIEF method was developed to evaluate charge heterogeneity over a wider range of AAV serotypes, demonstrating robustness and consistency in the results. Species variants seen in AAV serotypes may vary depending on the stability condition, peptide sequence, or manufacturing process.
- Distinctive changes in the fingerprint profile for each AAV serotype relating to %Total for each species variant could indicate instability leading to reduced potency and increased presence of post-translational modifications (PTMs).
- Acidic variants should be monitored during the experimentation to determine their impact on transduction.

MATERIALS & METHODS

- To measure charge heterogeneity of each AAV serotype, a cIEF method was developed.
- This method utilized the Maurice from ProteinSimple (San Jose, CA) to separate the charged variants of each AAV under a single method and set of running conditions.
- The method was evaluated using the International Council of Harmonisation Q2 guidelines specific to method development, which looked at linearity, precision, accuracy, and limit of quantification.
- Thermal stress conditions were imposed on each AAV consistent with chemical mobilization capillary specifically on accelerated, elevated, and high-temperature stress conditions.
- Oxidation stress treatment was another condition imposed to generate AAVs that have an increased abundance of oxidation PTMs confirmed by mass spectrometry.
- Each treated set of AAV was then run on a novel *in vitro* protein expression method to evaluate total resultant protein expression levels in reference to non-treated AAV.
- Descriptive statistics, one-way analysis of variance (ANOVA), and unpaired t-test were used for statistical analysis of the data.

RESULTS

METHODS EVALUATION AND PROFILES

Each AAV serotype evaluated using this method resulted in a reproducible cIEF profile with consistent %Total species variants. Each AAV serotype had comparable isoelectric point (pI) values of the main peak species, but **Figure 1B** shows some variability between each, while the internal standard pI markers are consistent in each.

Figure 1. AAV Serotype Profiles

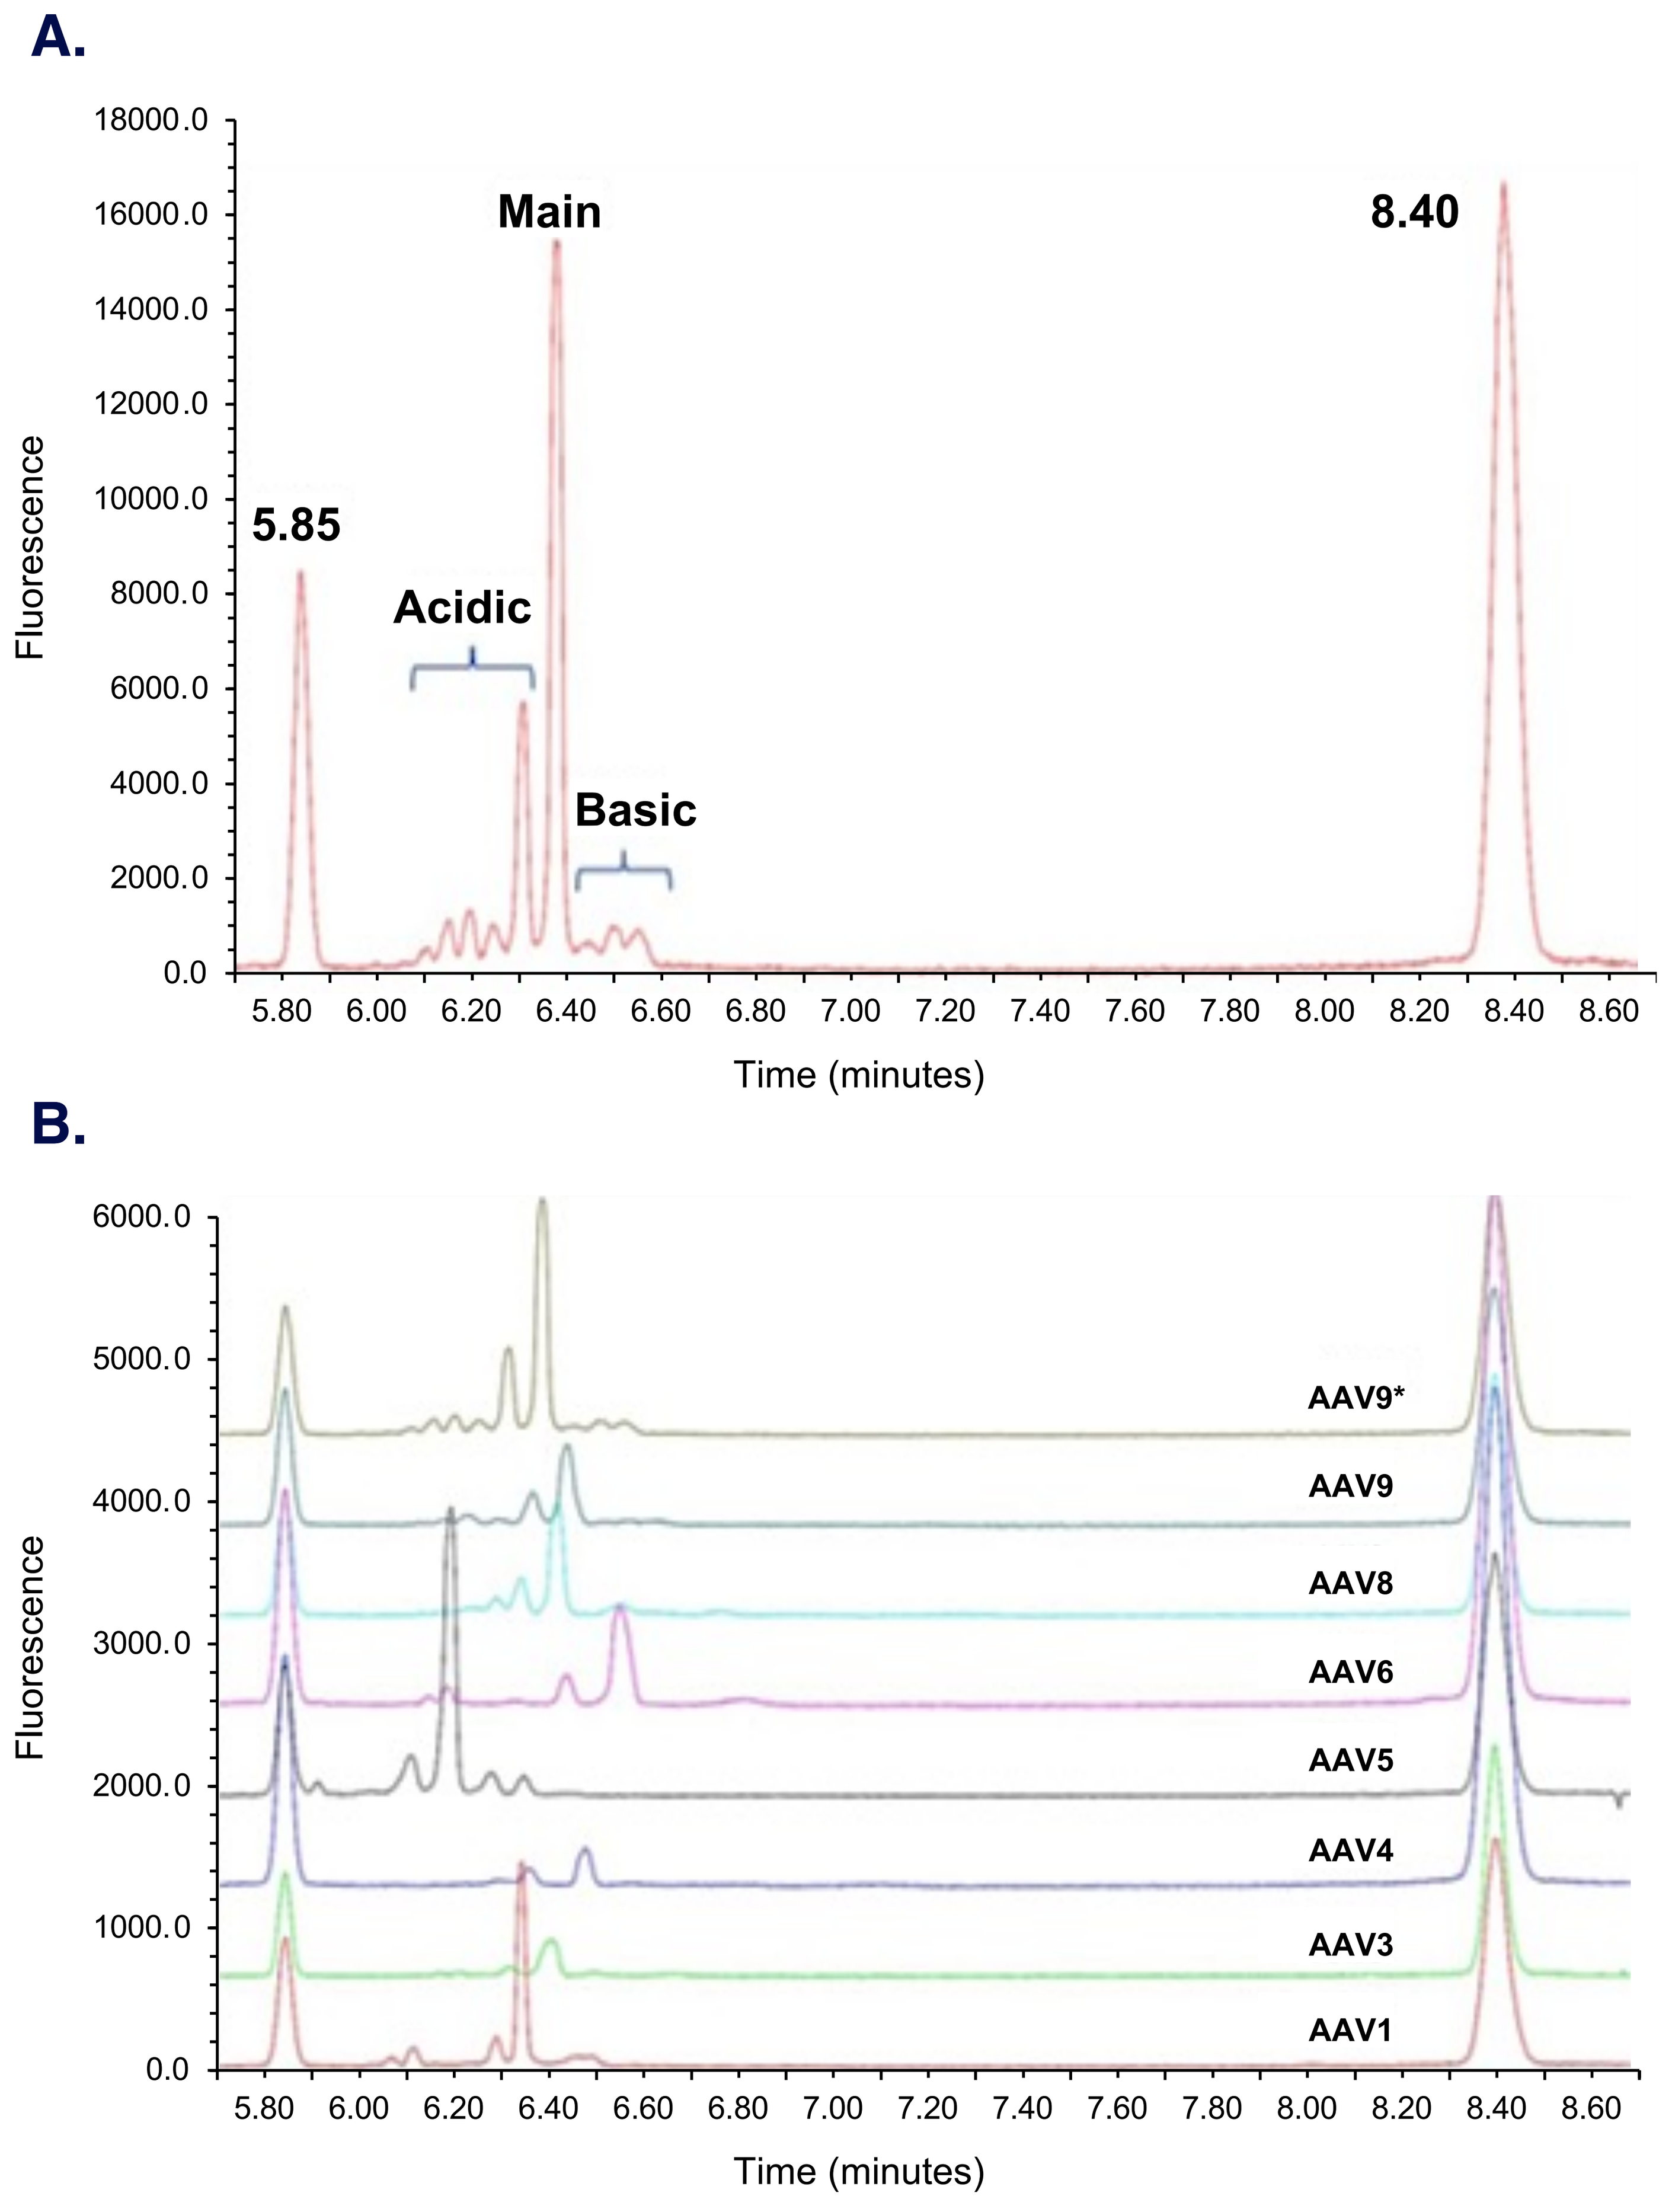
A.
Fluorescence
Time (minutes)
Main
8.40
5.85
Acidic
Basic
B.
AAV9*
AAV9
AAV8
AAV6
AAV5
AAV4
AAV3
AAV1

A) AAV9 overlay demonstrating the separation of the three primary species variants.
B) A variety of different AAV serotypes run under the same method conditions.

*Empty fraction only.

THERMAL AND OXIDATIVE IMPACT ON AAV

Each AAV was subjected to thermal stress under accelerated, elevated, and high-temperature stress and demonstrated changes in species variants and shifts in the main peak pI. Refer to **Figure 2** for the electropherogram and %Total species results for AAV9. Each AAV was subjected to oxidative stress conditions that induced a higher abundance of peptides containing the oxidation modification. Refer to **Figure 3** for oxidative-stress results for AAV9. These data also represent the change in %Total species by cIEF and diminished level of protein expression compared to untreated AAV9.

Figure 2. AAV9 Thermal Stress Impact on Charge Heterogeneity and Resultant Protein Expression

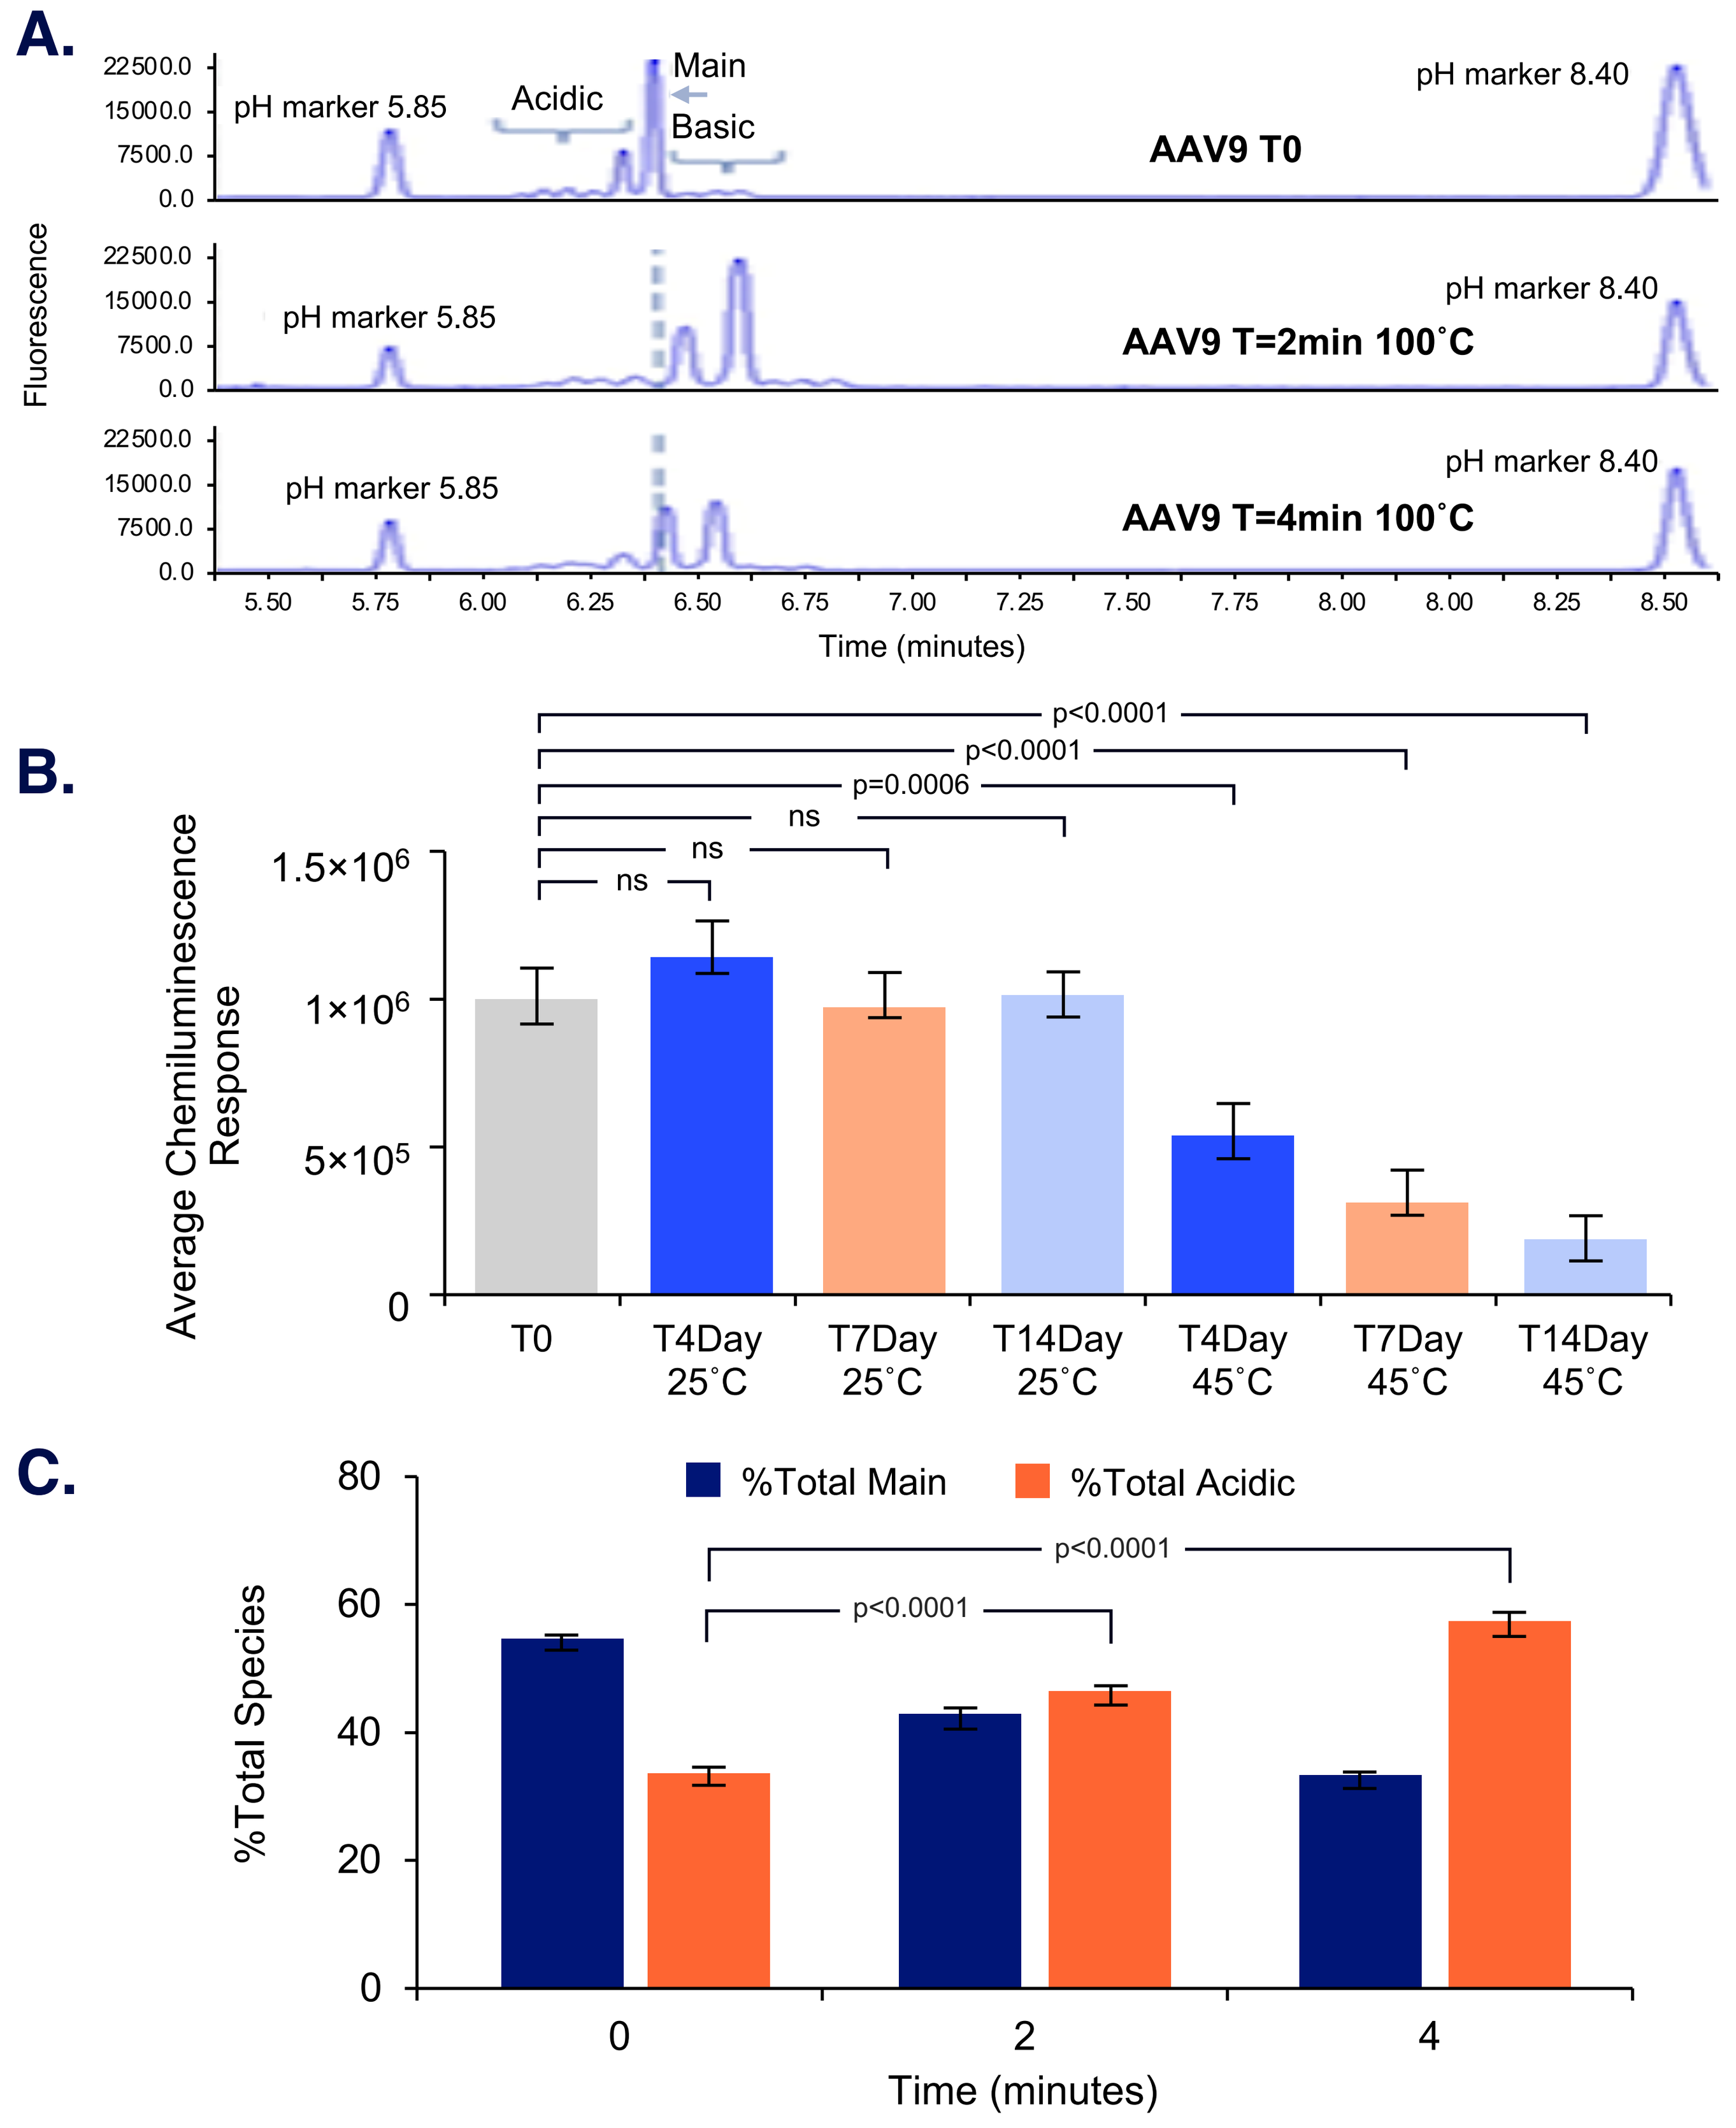
A.
pH marker 5.85
Acidic
Main
Basic
pH marker 8.40
AAV9 T0
AAV9 T=2min 100˚C
AAV9 T=4min 100˚C
Fluorescence
Time (minutes)
B.
Average Chemiluminescence Response
p<0.0001
p<0.0001
p=0.0006
ns
ns
ns
T0
T4Day 25˚C
T7Day 25˚C
T14Day 25˚C
T4Day 45˚C
T7Day 45˚C
T14Day 45˚C
C.
%Total Species
%Total Main
%Total Acidic
p<0.0001
p<0.0001
Time (minutes)

A) The cIEF electropherogram demonstrated a shift in main peak pI at high-temperature stress. **B)** Protein expression levels at the highest multiplicity of infection showed most variability in the 45°C condition compared to the control, p<0.0001. **C)** The %Total for Acidic species in each condition increased compared to the control, p<0.0001.

Figure 3. AAV9 Oxidative Stress Impact on Charge Heterogeneity and Resultant Protein Expression

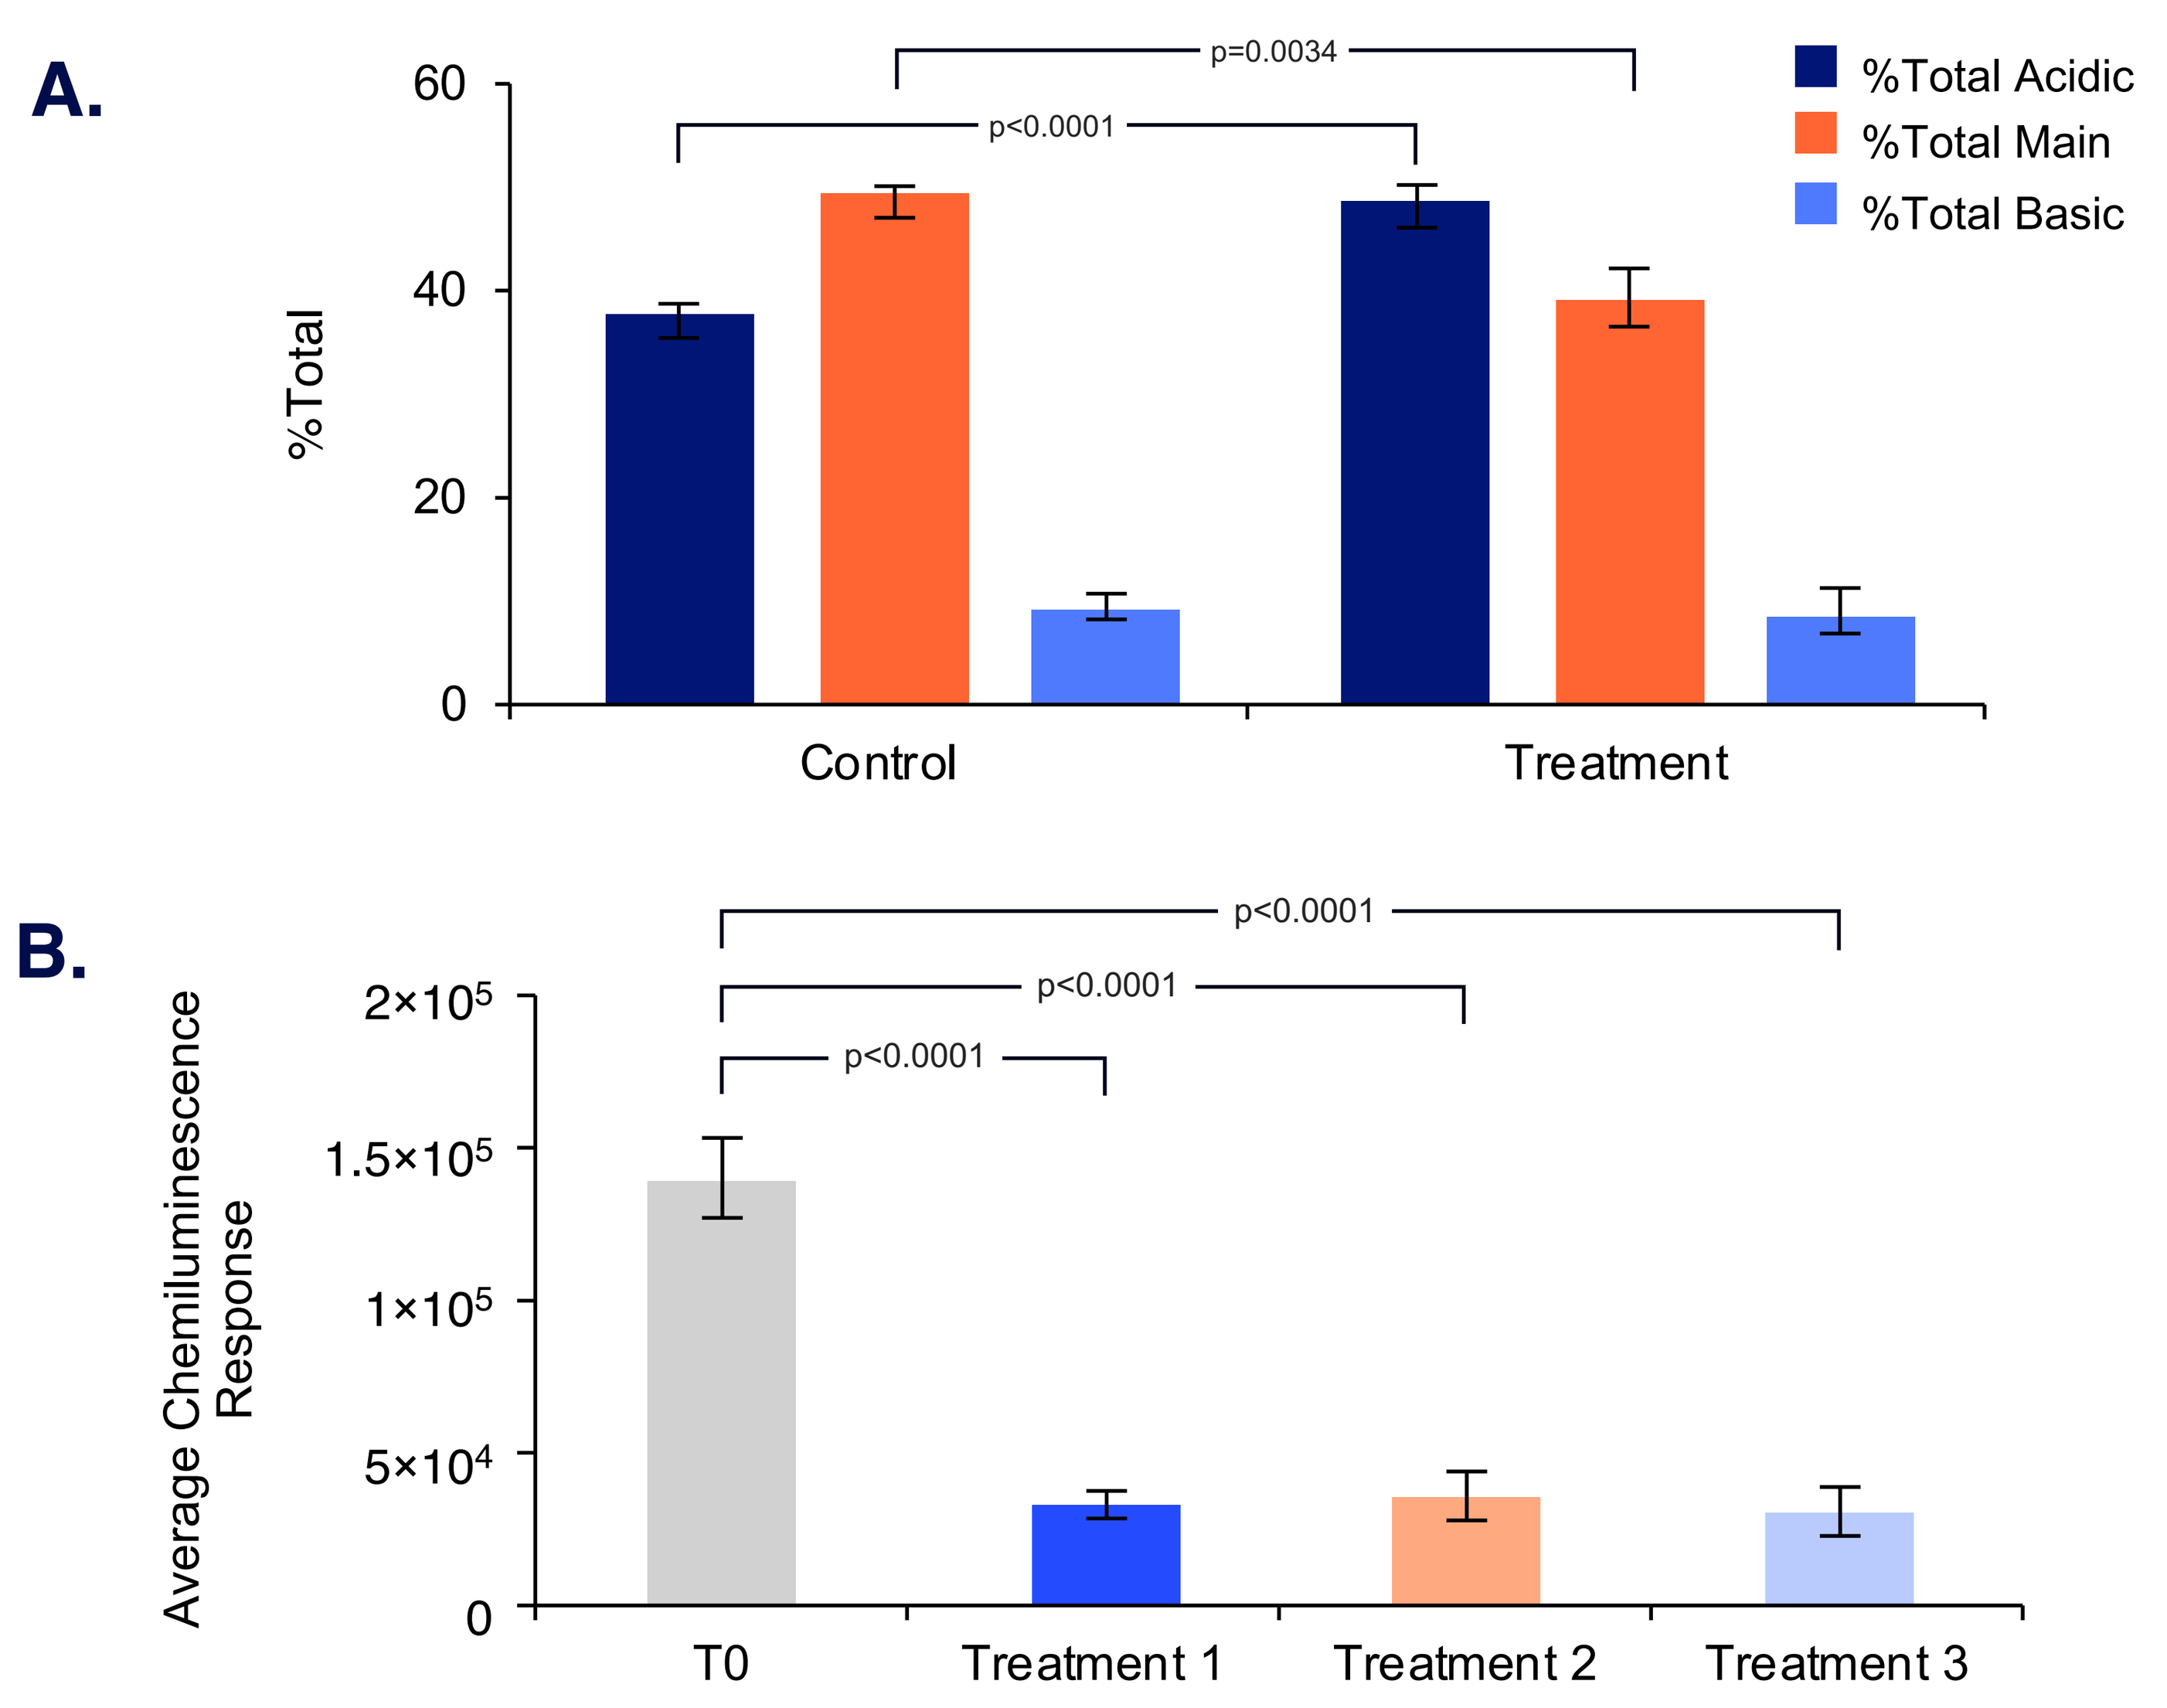
A.
%Total
p=0.0034
p<0.0001
%Total Acidic
%Total Main
%Total Basic
Control
Treatment
B.
Average Chemiluminescence Response
p<0.0001
p<0.0001
p<0.0001
T0
Treatment 1
Treatment 2
Treatment 3

A) Oxidation treatment samples showed an increase in %Total for acidic variants compared to the control, p<0.0001, whereas there is a decrease in %Total for main species variants.
B) Resultant protein expression was diminished in the same oxidation treatment samples compared to the control AAV9, p<0.0001.

CONCLUSIONS

This platform cIEF method demonstrated its capabilities to robustly characterize each of the AAV serotypes analyzed. The correlation between %Total species variants was apparent when focusing on the change in %Total for acidic and %Total for main species variants vs. resultant protein expression levels. Our data demonstrate that charge heterogeneity variability arising from PTMs influenced how AAV can deliver GOI to target cells resulting in decreased levels of protein expression. Uniform charge profiles were maintained in AAV when kept under proper storage conditions (e.g., -80˚C), while stress conditions (e.g., thermal) did not promote increased protein expression. With each successive time-hold stress condition, potency was decreased, which could have been due to the lower number of full capsids present, capsid aggregation, or receptor recognition impact. These findings underscore how charge heterogeneity can be a predictor of resultant protein expression depending on capsid stability.

Presented at the ASGCT 28th Annual Meeting, May 13-17, 2025, New Orleans, LA, USA.